2318 AASLD The Liver Meeting® Nov. 4-8, 2022

ID119031166, A Novel Potent Non-bile acid FXR Agonist, Improves NASH and Liver Fibrosis Without Plasma LDL-C Elevation in a Diet-induced Obese NASH Hamster Model

An-Na Moon[1], Yoonsuk Lee[1], Dong-Keun Song[1], François Briand[2]

[1]*iLeadBMS Co., Ltd., Hwaseong, South Korea,* [2]*Physiogenex, Escalquens, France*

iLeadBMS

Background and Aim

Farnesoid X Receptor (FXR), mainly expressed in liver and intestine, has been identified as a promising drug target for the treatment of non-alcoholic steatohepatitis (NASH). Several non-bile acid FXR agonists have been tested in clinical trials for NASH but the unwanted side effects (e.g., LDL-C increase) should be mitigated. Here, we report on the benefits of ID119031166 as a novel, potent and non-bile acid FXR agonist in the diet-induced obese NASH hamster, a preclinical model with human-like NASH and lipoprotein cholesterol metabolism.

Methods

- In vitro activities of FXR and other nuclear receptors were determined by reporter assays. TGR5 activity was measured by cell-based cAMP assay.
- In primary human hepatocytes, the induction of FXR target gene SHP by OCA or ID119031166 was examined by qPCR after 24 h incubation.
- After daily oral administration of 3, 10 and 30 mg/kg for 14 days in normal hamsters (n=6), FXR target engagement in liver and ileum was determined by measuring FXR target gene expression. Plasma PK parameters at 10 and 30 mg/kg were also assessed in the hamsters. Liver and ileum tissues in the hamsters were collected at 2 h post-dose to determine tissue/plasma ratio.
- The efficacy of ID119031166 on NASH and liver fibrosis was evaluated in a diet-induced obese NASH hamster model. Following 5 weeks of oral treatment with either vehicle or ID119031166 30 mg/kg/day (n=15), liver histology and plasma biochemistry were performed.

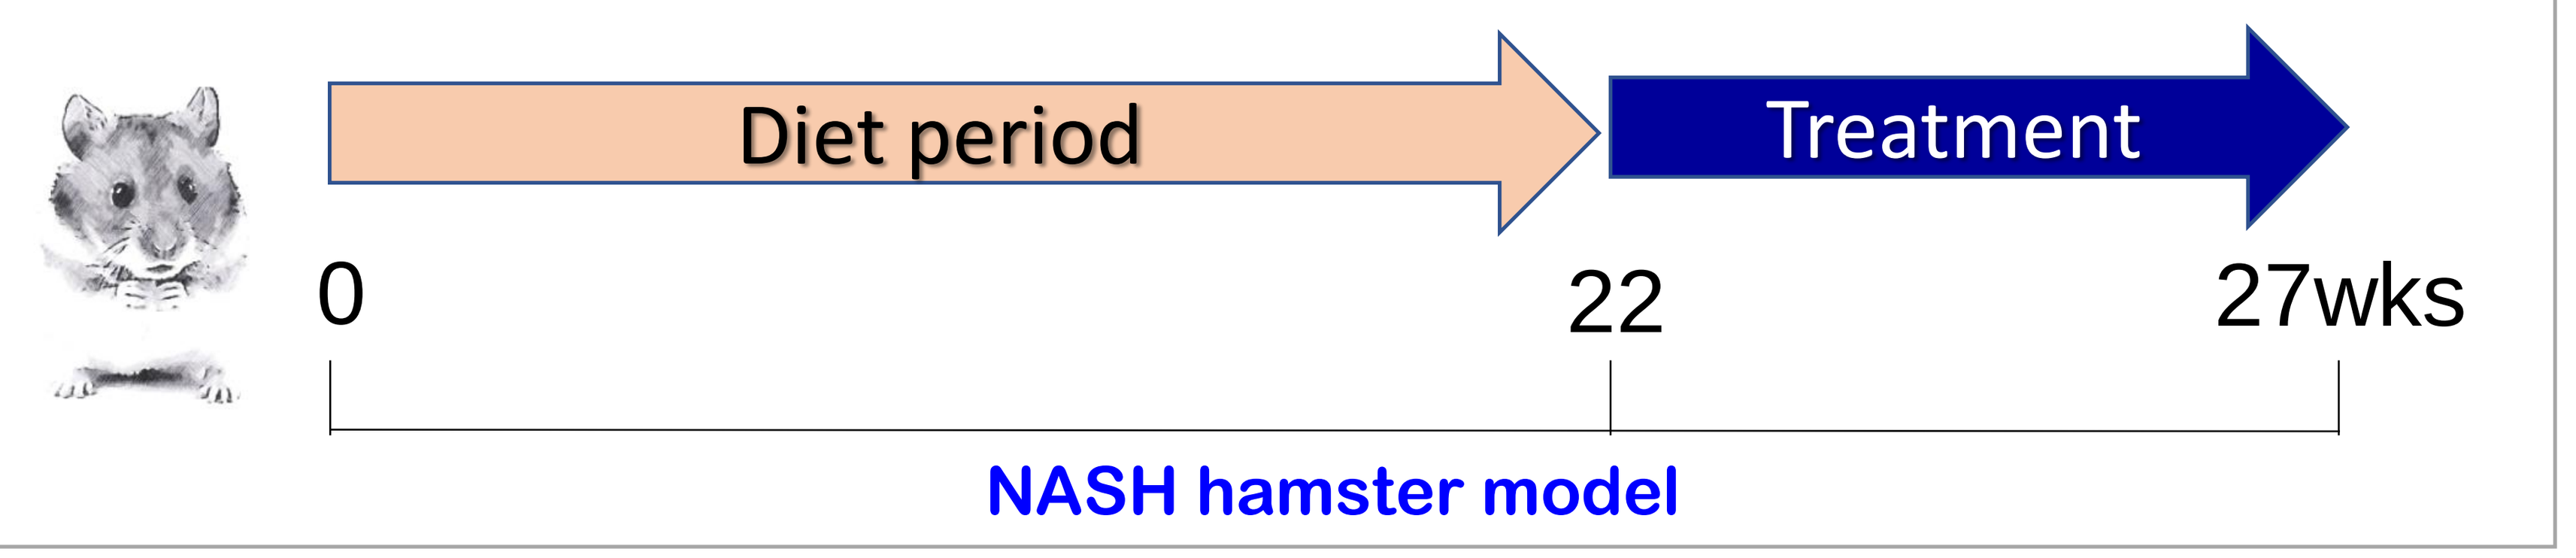
Diet period
Treatment
0
22
27wks
NASH hamster model

Results

1. In vitro activity

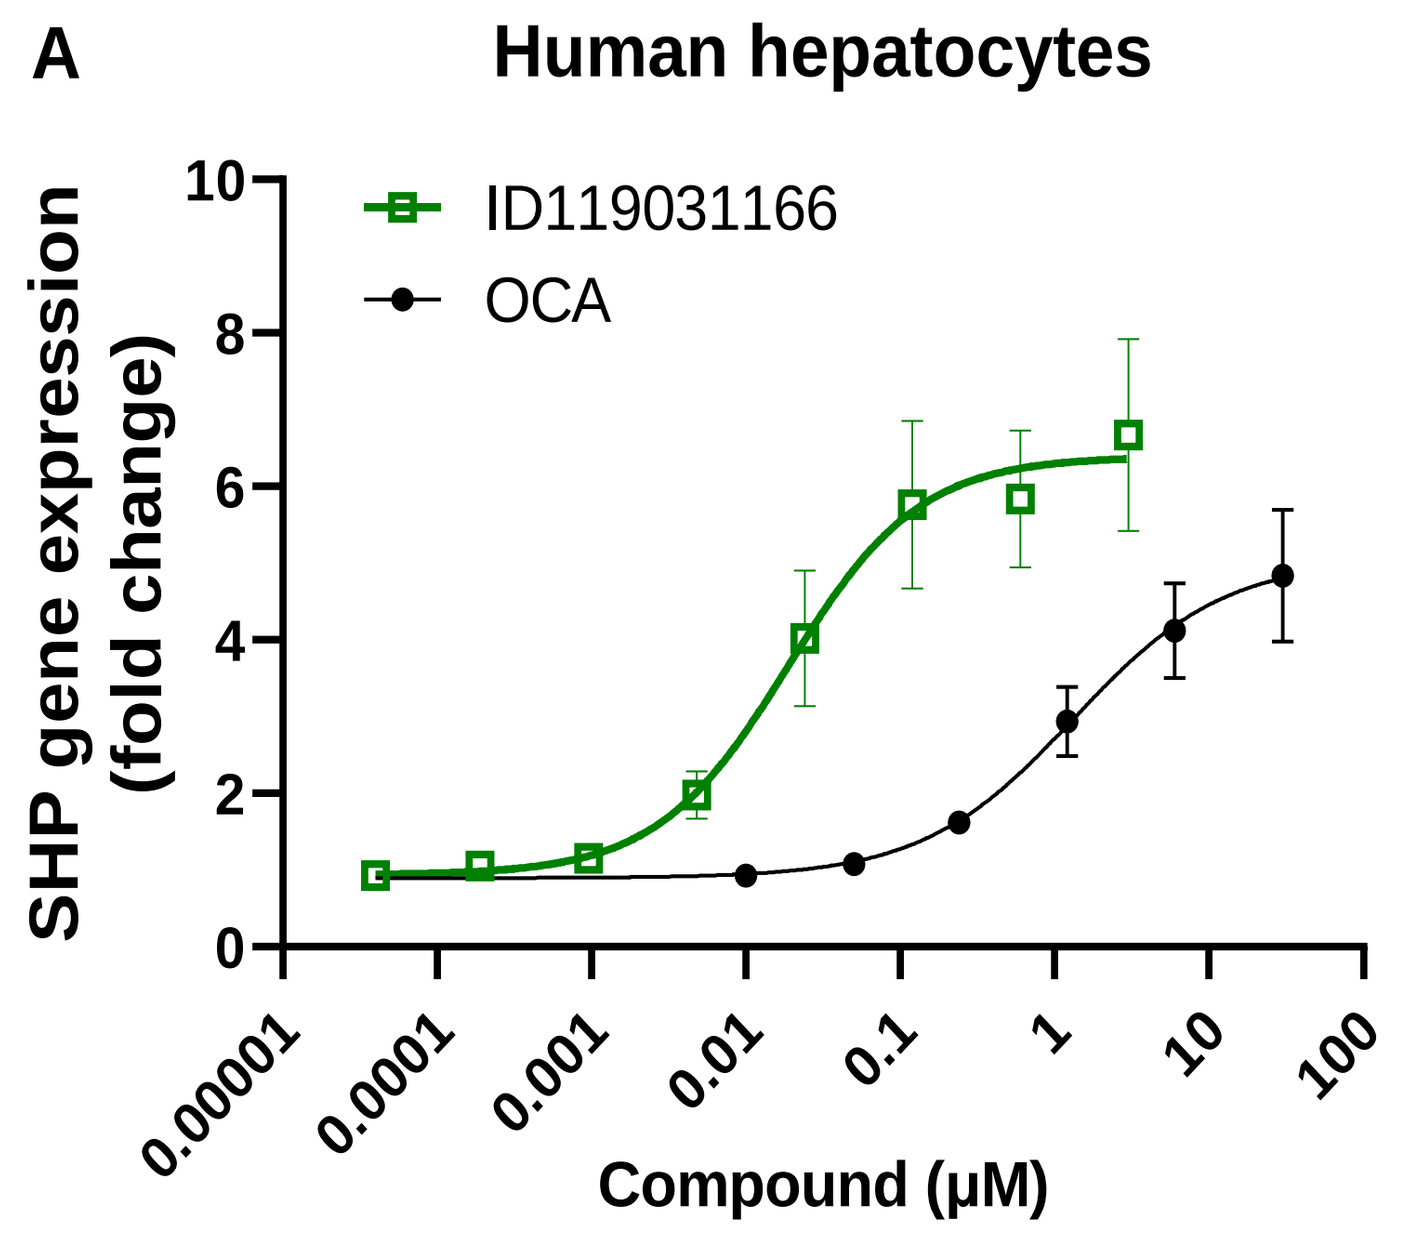
A
Human hepatocytes
ID119031166
OCA
SHP gene expression (fold change)
Compound (μM)

Assay	ID119031166
Human FXR	EC_{50} 5 nM
Hamster FXR	EC_{50} 24 nM
Human TGR5	No activity up to 200uM
Nuclear receptor panel	No activity up to 30uM

- ID119031166 is a potent FXR agonist with high selectivity against other nuclear receptors and TGR5.

2. 14-day PK/PD study in hamsters

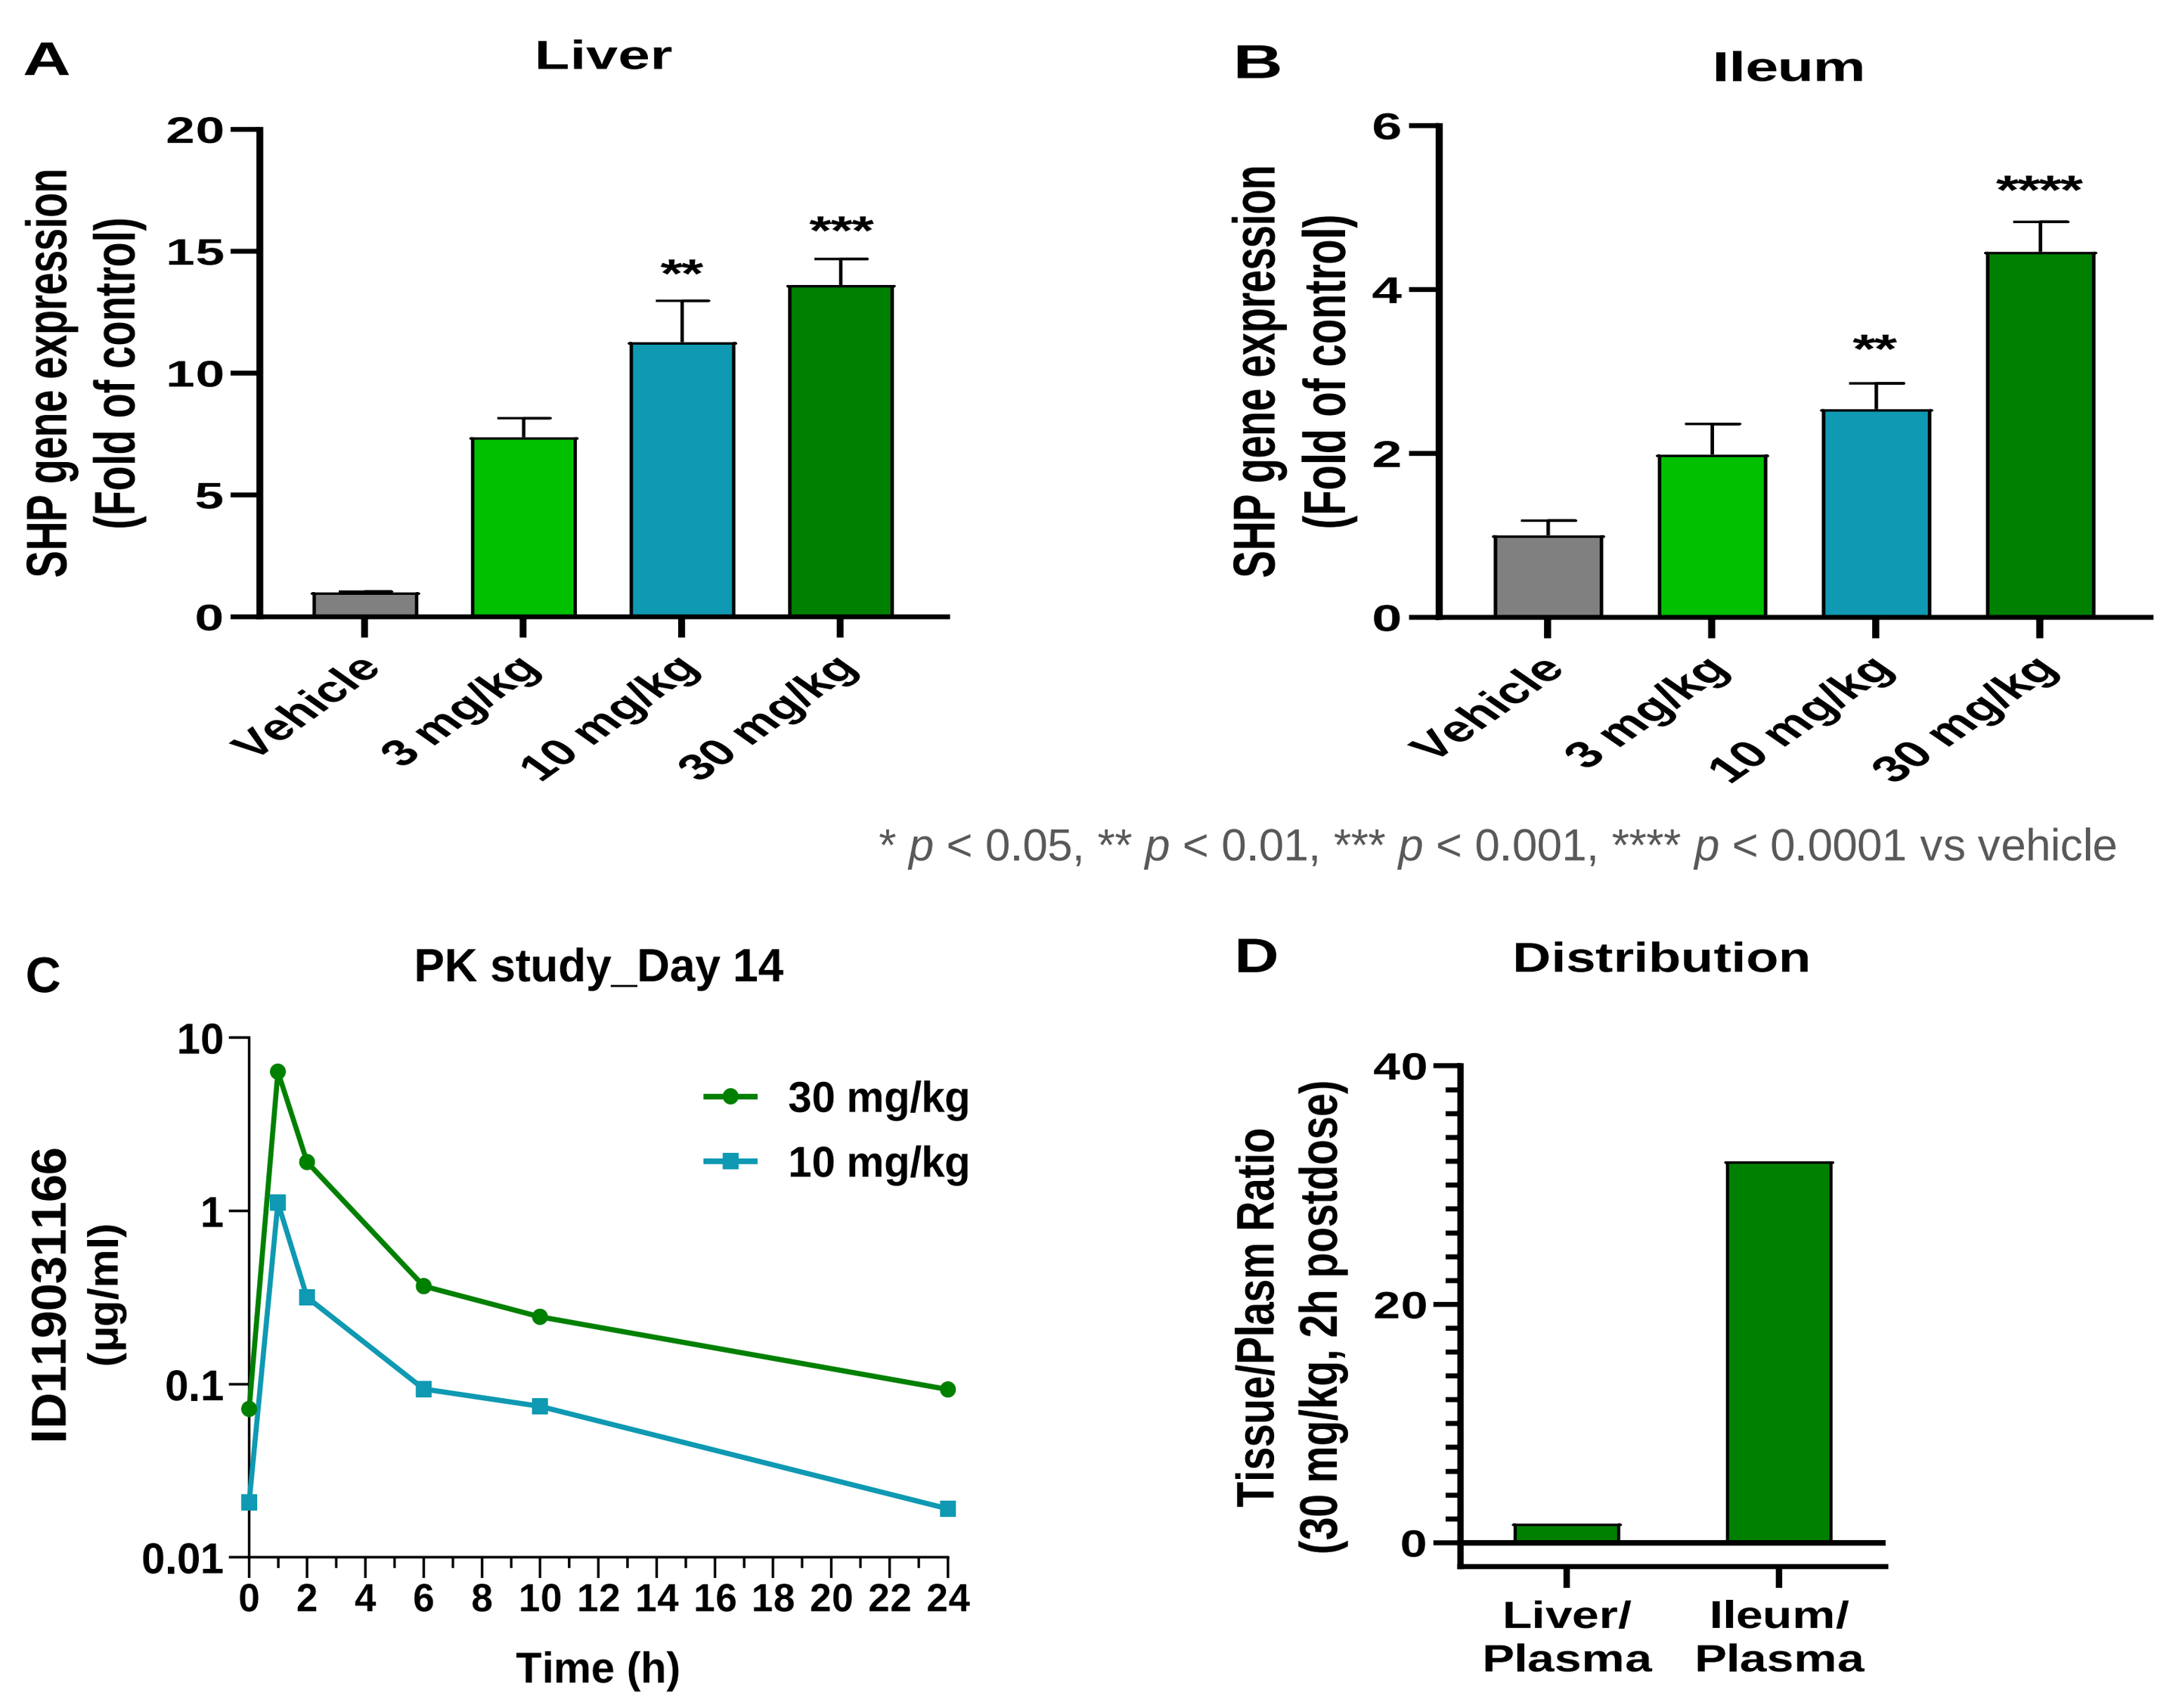
A
Liver
SHP gene expression (Fold of control)
Vehicle
3 mg/kg
10 mg/kg
30 mg/kg
B
Ileum
C
PK study_Day 14
30 mg/kg
10 mg/kg
ID119031166 (μg/ml)
Time (h)
D
Distribution
Tissue/Plasm Ratio (30 mg/kg, 2h postdose)
Liver/ Plasma
Ileum/ Plasma

* *p* < 0.05, ** *p* < 0.01, *** *p* < 0.001, **** *p* < 0.0001 vs vehicle

- SHP gene expression in liver and ileum at 2 h post-dose was dose-dependently induced up to 14-fold, relative to vehicle.
- Plasma exposure in the hamsters increased with dose and C_{max} and AUC_{0-24} values at 30 mg/kg were 6.4 μg/mL and 15.5 h·μg/mL, respectively. Interestingly, preferential distribution to the ileum than the liver was also shown (Ileum-to-plasma ratio > 30) at 30 mg/kg.

3. Efficacy (Diet-induced Obese NASH Hamster model)

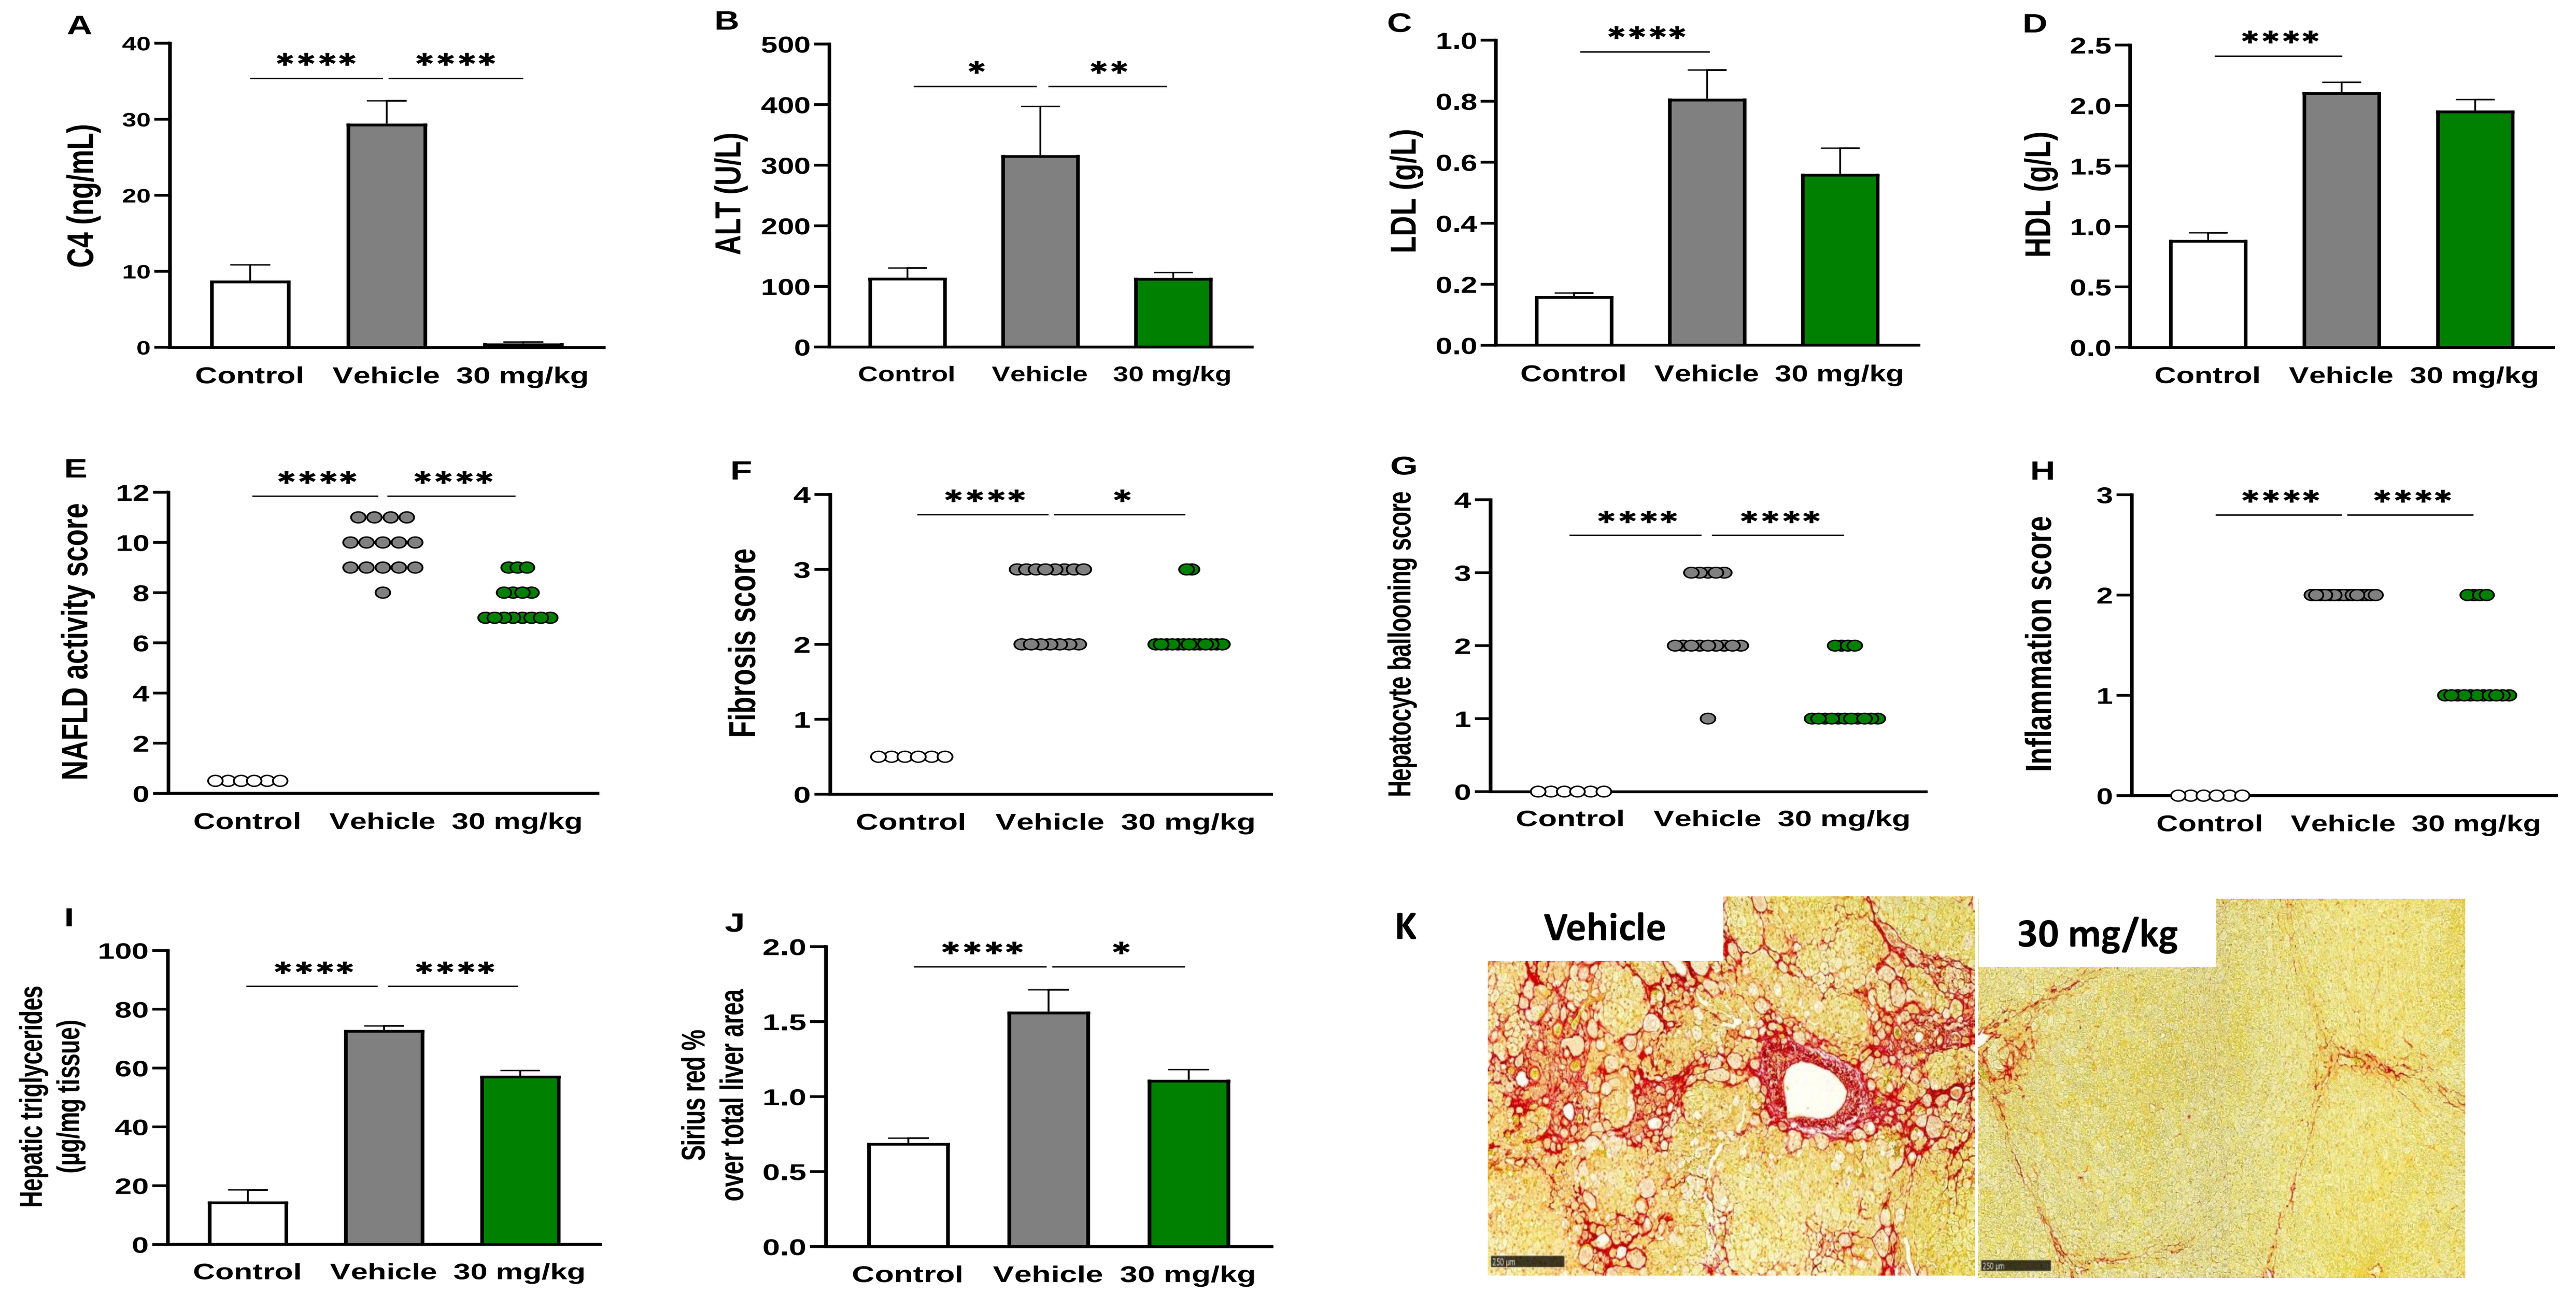
A
C4 (ng/mL)
B
ALT (U/L)
C
LDL (g/L)
D
HDL (g/L)
E
NAFLD activity score
F
Fibrosis score
G
Hepatocyte ballooning score
H
Inflammation score
I
Hepatic triglycerides (μg/mg tissue)
J
Sirius red % over total liver area
Control
Vehicle
30 mg/kg
K
Vehicle
30 mg/kg

* *p* < 0.05, ** *p* < 0.01, **** *p* < 0.0001

- In the diet-induced obese NASH hamster model, ID119031166 30 mg/kg significantly reduced plasma C4 (98%, p<0.0001 vs vehicle) and ALT (64%, p<0.01) levels and showed significant improvement in total NAFLD activity score(p<0.0001), including lower fibrosis(p<0.05), inflammation (p<0.0001) and hepatocyte ballooning (p<0.0001) scores. In line with the fibrosis score, % Sirius Red labelling was significantly reduced (p<0.05).
- ID119031166 at 30 mg/kg led to significant reduction of hepatic triglycerides (p<0.0001).
- Importantly, these benefits were observed with no changes in plasma HDL-C and LDL-C levels.

Conclusion

ID119031166, a novel, potent, selective and non-bile FXR agonist, demonstrates efficacy in a diet-induced obese NASH hamster model with no changes in LDL-C and HDL-C levels. These preclinical results support further clinical investigation of ID119031166. Ildong Pharmaceutical initiates a US-based phase 1 clinical trial of ID119031166 in healthy volunteers.

Contact : anna.moon@ileadbms.com

Disclosure

An-Na Moon and Dong-Keun Song are employees and stock shareholders of iLeadBMS Co., Ltd. Yoonsuk Lee is a board member and stock shareholder of iLeadBMS Co., Ltd. François Briand is an employee of Physiogenex (a preclinical Contract Research Organization) and has shares in the company. iLeadBMS Co., Ltd. has received research funding from Ildong Pharmaceutical Co., Ltd.

© 2022 Ildong Pharmaceutical Co., Ltd. All rights reserved