Lanifibranor improves NASH, fibrosis and diastolic dysfunction in a hamster preclinical model of diet induced NASH

Wettstein G[1], Briand F[2], Sulpice T[2], Junien JL[1], Broqua P[1]

[1] INVENTIVA, Daix, France. [2] Physiogenex, Escalquens, France

1-INTRODUCTION

Lanifibranor is a well-balanced agonist of the 3 PPAR isotypes with anti-inflammatory and anti-fibrotic effects in pre-clinical models of NASH. The NATIVE phase 2b trial (NCT03008070) in non-cirrhotic NASH patients demonstrated beneficial effects of lanifibranor treatment on several histological endpoints including NASH resolution and improvement of fibrosis. It has been reported in several studies that NASH increases the risk for cardiovascular diseases. Moreover, patients with NASH are at higher risk of developing diastolic dysfunction. We evaluated in this study the potential of lanifibranor in preventing metabolic changes and diastolic dysfunction in a preclinical model of NASH, in comparison with the PPARγ agonist pioglitazone.

2-Material/Methods

Golden syrian Hamsters under free choice diet (free choice between a chow diet with normal tap water or a high fat/cholesterol diet (Safe Diets) with 10% fructose enriched tap water) for a period of 15 weeks developed NASH, fibrosis and diastolic dysfunction compared to control hamster under chow diet. Hamsters were then treated for a period of 5 weeks with either vehicle, lanifibranor 30mg/kg or pioglitazone 30mg/kg. At the end of the treatment, liver histology, genes expressions and biochemical analysis were performed. Diastolic dysfunction was evaluated by echocardiography and defined as an absence of change in left ventricular ejection fraction, an increase in E/A and E/E' ratio and a decrease in E'/A' ratio as well as in isovolumic relaxation time (IVRT).

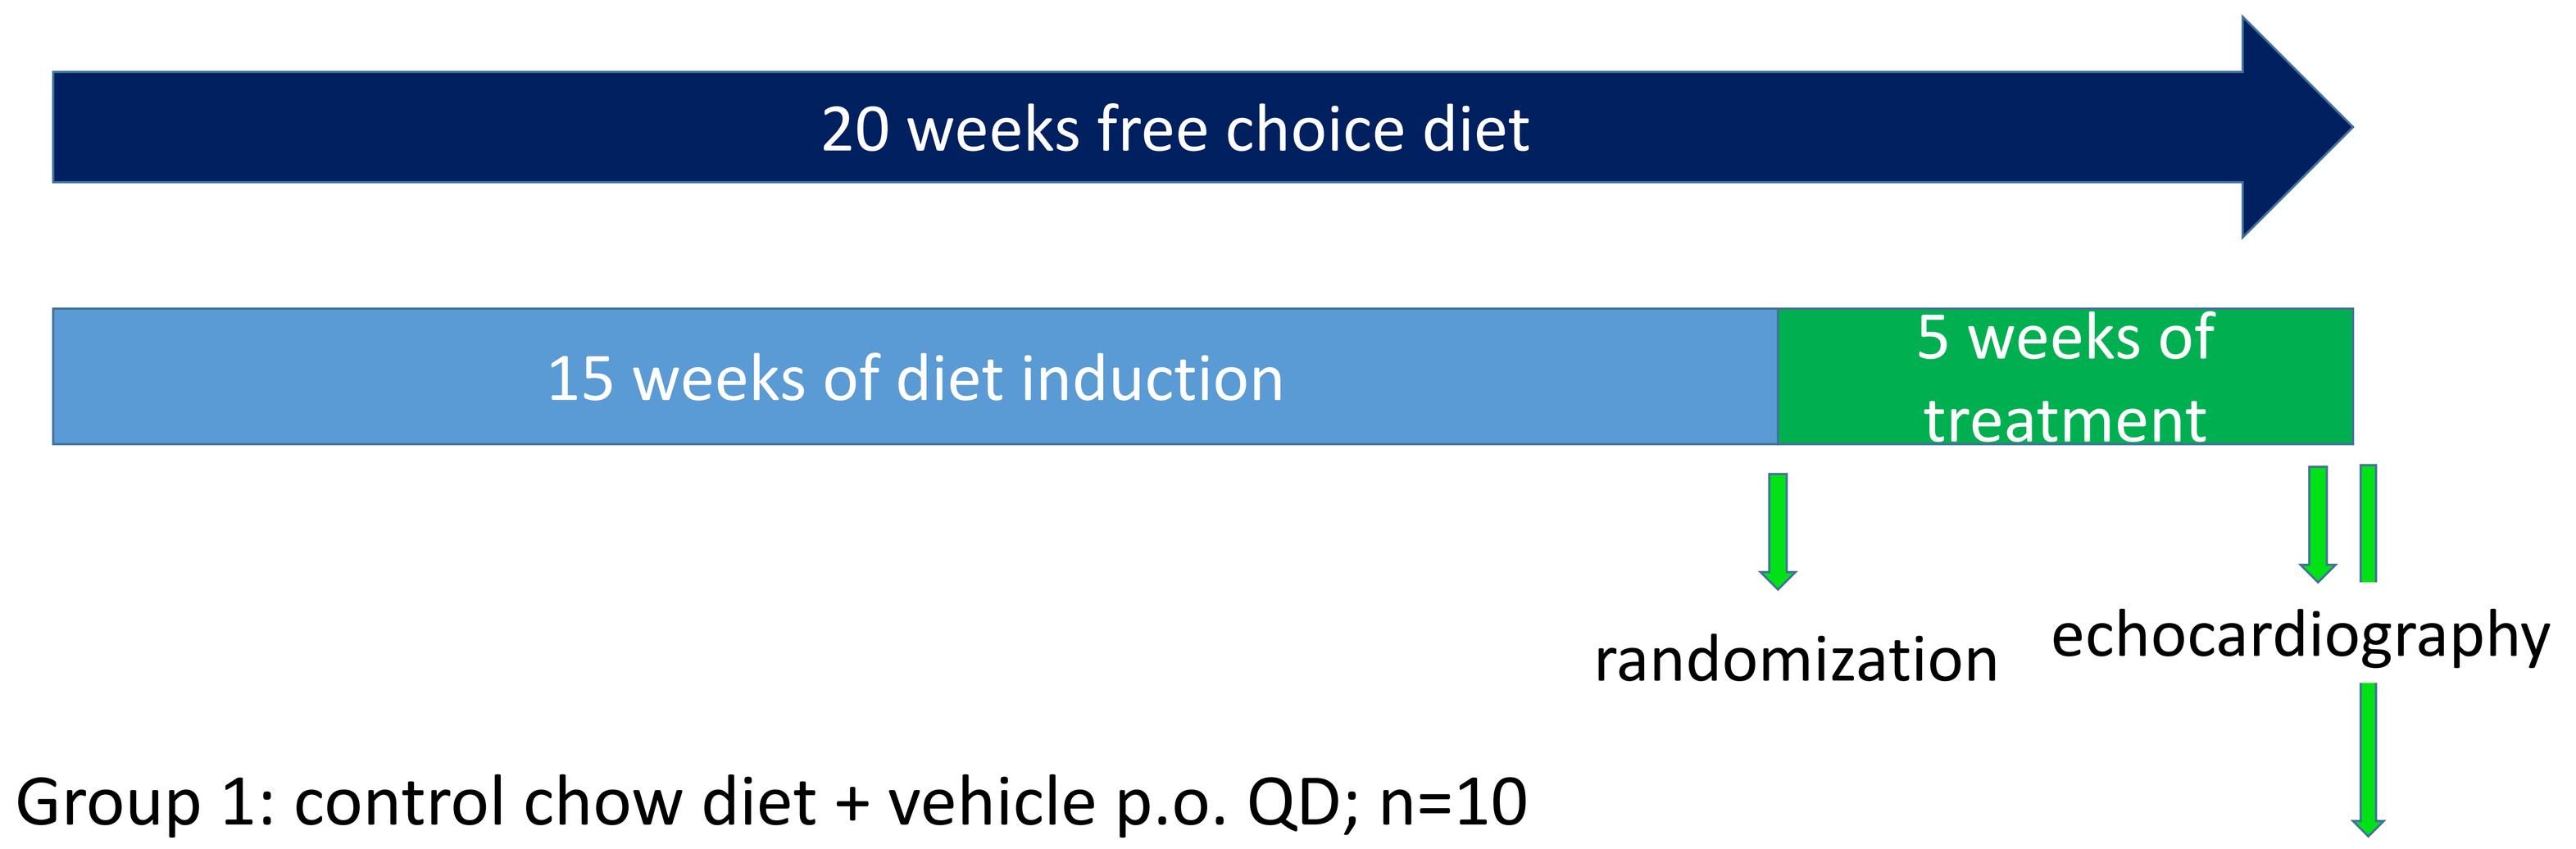
20 weeks free choice diet
15 weeks of diet induction
5 weeks of treatment
randomization
echocardiography
Blood and liver collection

Group 1: control chow diet + vehicle p.o. QD; n=10
Group 2: Free choice diet + vehicle p.o. QD; n=10
Group 3: Free choice diet + lanifibranor p.o. QD; n=10
Group 4: Free choice diet + pioglitazone p.o. QD; n=10

3-RESULTS

* Vs vehicle with * p<0.05; **p<0.01; ***p<0.001

Metabolic parameters: glucose and lipids

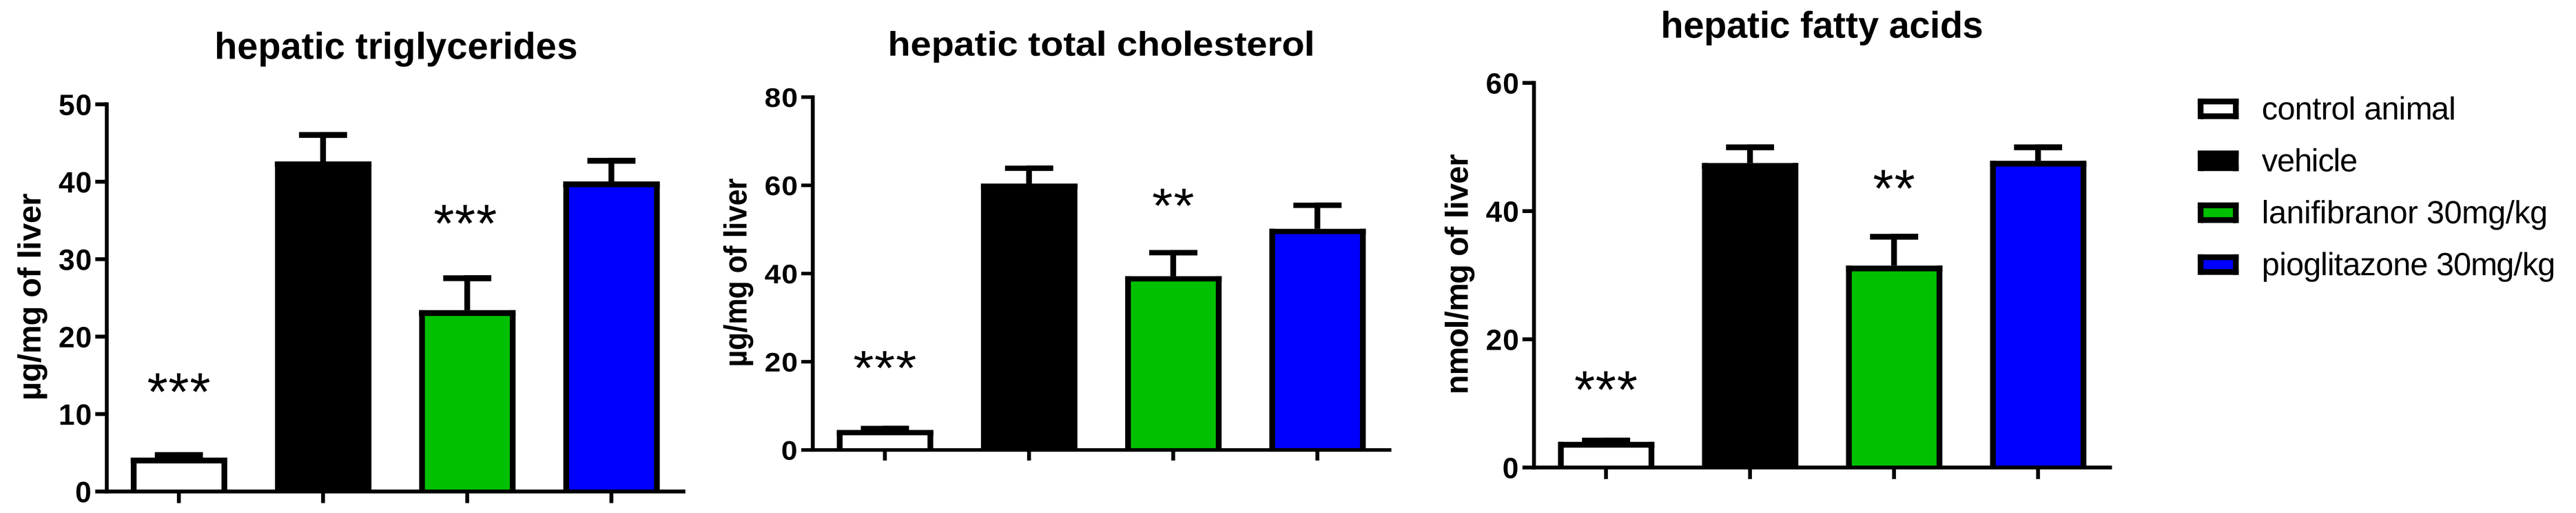
hepatic triglycerides
µg/mg of liver
hepatic total cholesterol
µg/mg of liver
hepatic fatty acids
nmol/mg of liver
control animal
vehicle
lanifibranor 30mg/kg
pioglitazone 30mg/kg

Lanifibranor, but not pioglitazone, reduced hepatic cholesterol, hepatic triglycerides and hepatic fatty acids increase due to High Fat Diet.

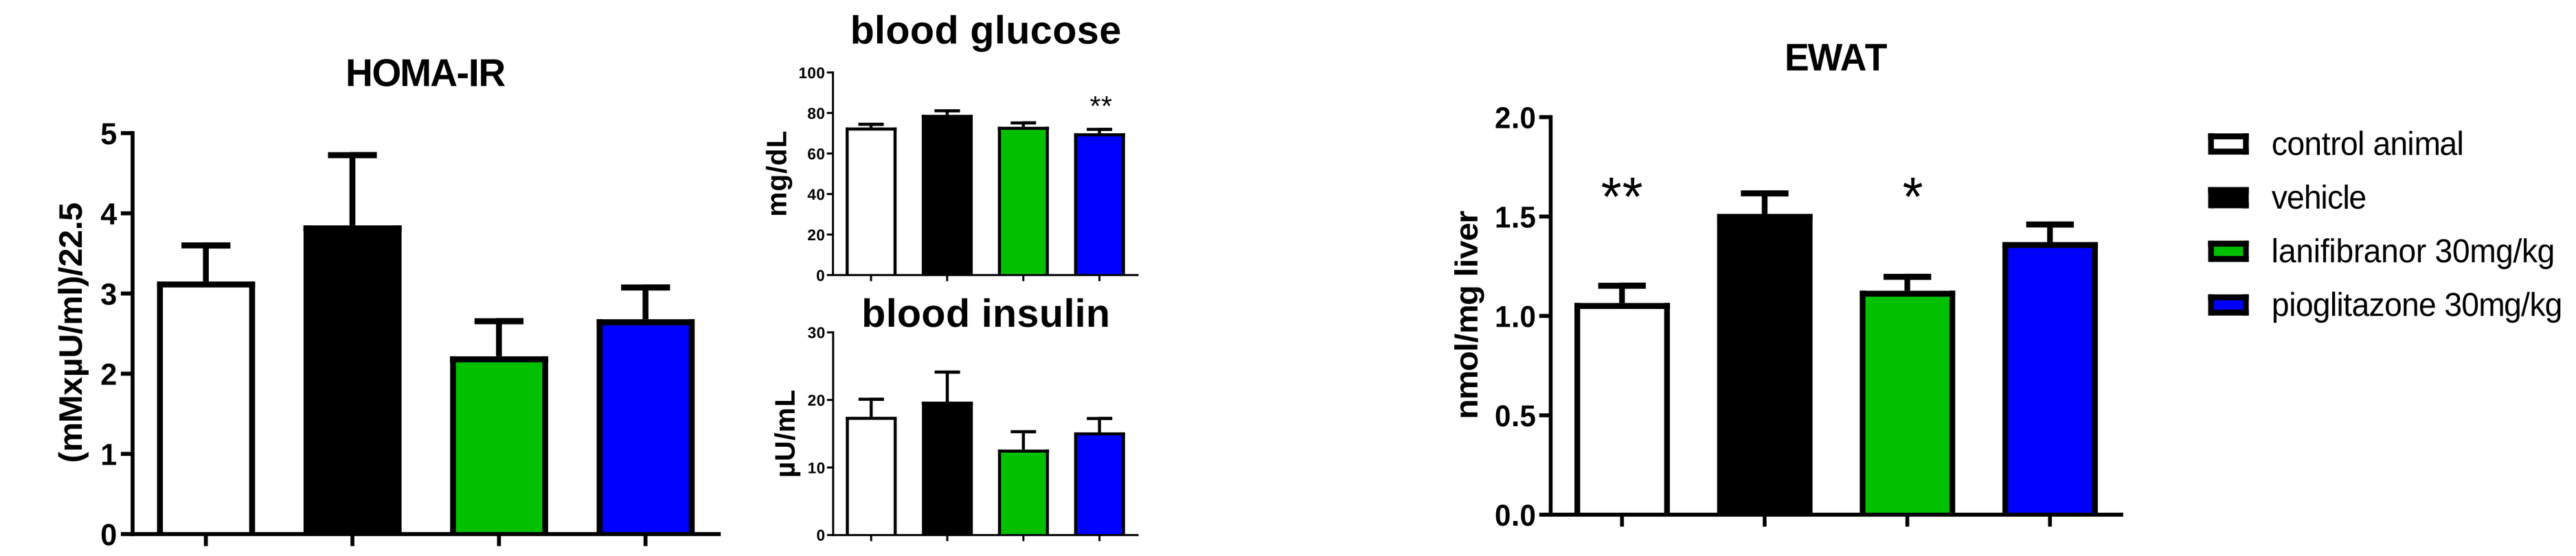
HOMA-IR
(mMxµU/ml)/22.5
blood glucose
mg/dL
blood insulin
µU/mL
EWAT
nmol/mg liver
control animal
vehicle
lanifibranor 30mg/kg
pioglitazone 30mg/kg

Lanifibranor and pioglitazone tend to reduced blood glucose, blood insulin and the resultant HOMA-IR index.

Lanifibranor, but not pioglitazone, significantly reduced epididymal white adipose tissue increase due to High Fat Diet.

NASH features and fibrosis

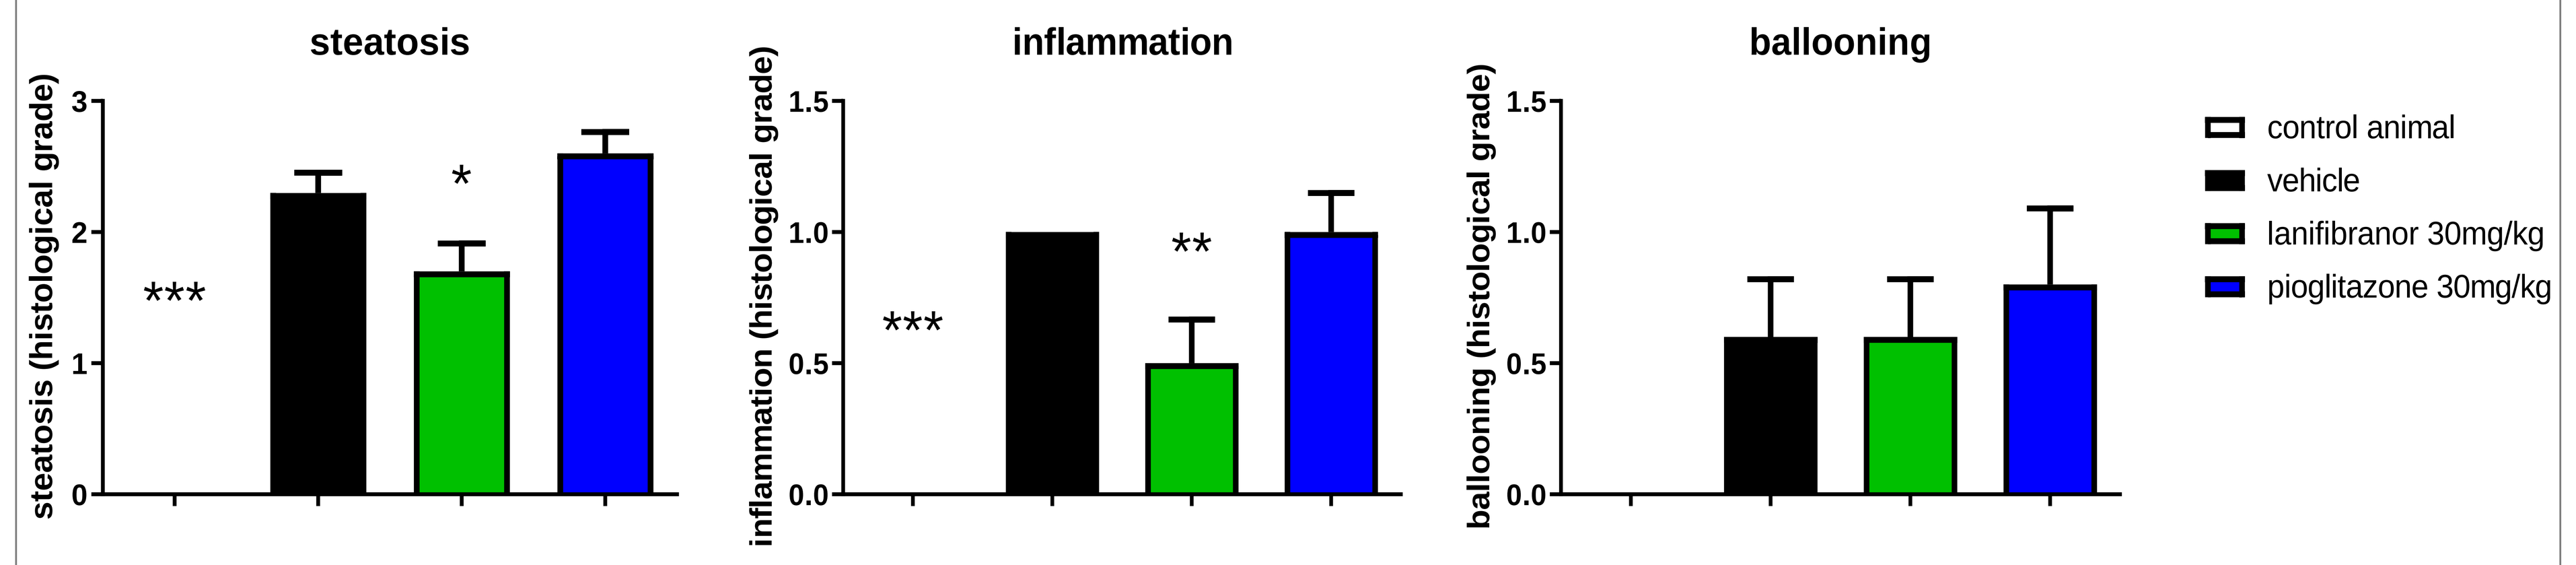
steatosis
steatosis (histological grade)
inflammation
inflammation (histological grade)
ballooning
ballooning (histological grade)
control animal
vehicle
lanifibranor 30mg/kg
pioglitazone 30mg/kg

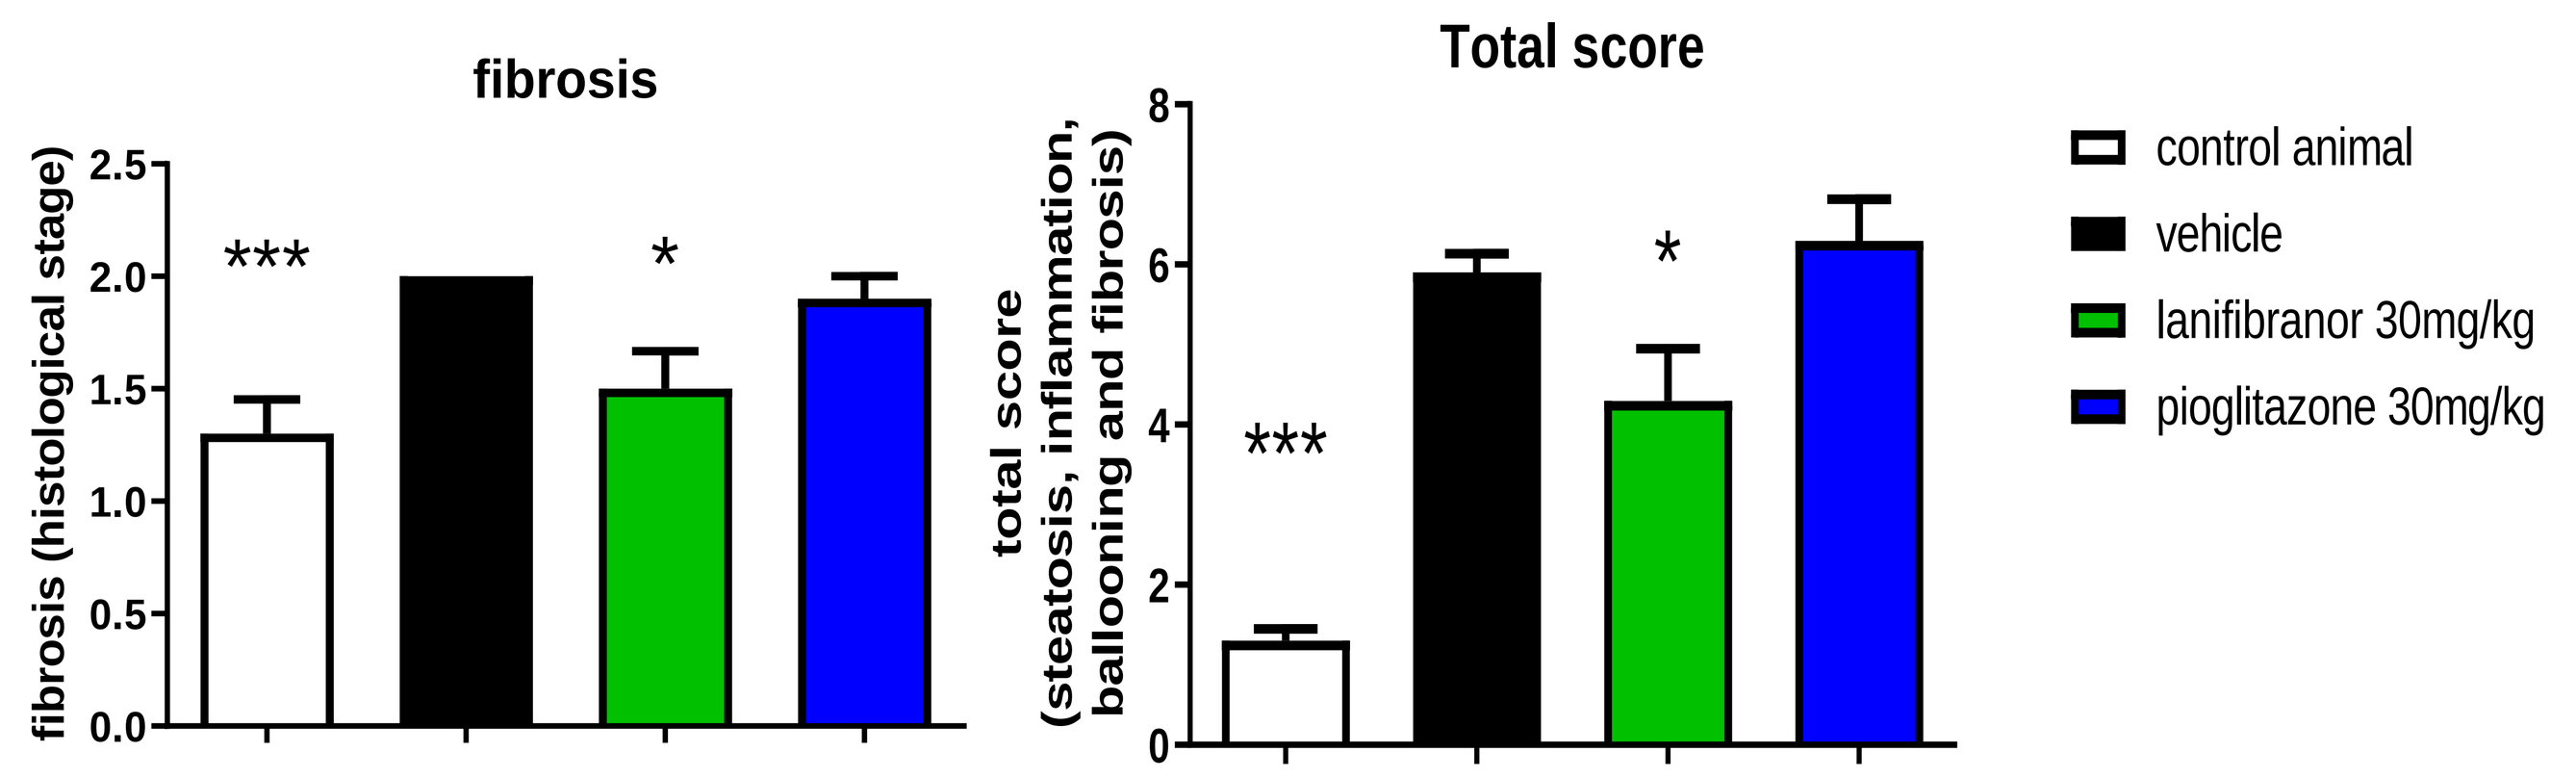
fibrosis
fibrosis (histological stage)
Total score
total score (steatosis, inflammation, ballooning and fibrosis)
control animal
vehicle
lanifibranor 30mg/kg
pioglitazone 30mg/kg

High Fat diet produced a significant increase in steatosis, liver inflammation and fibrosis but not ballooning.

Lanifibranor, but not pioglitazone, significantly decreased steatosis, inflammation, fibrosis and consequently the total score taking into account the 3 NASH features and fibrosis.

Inflammatory and fibrosis genes

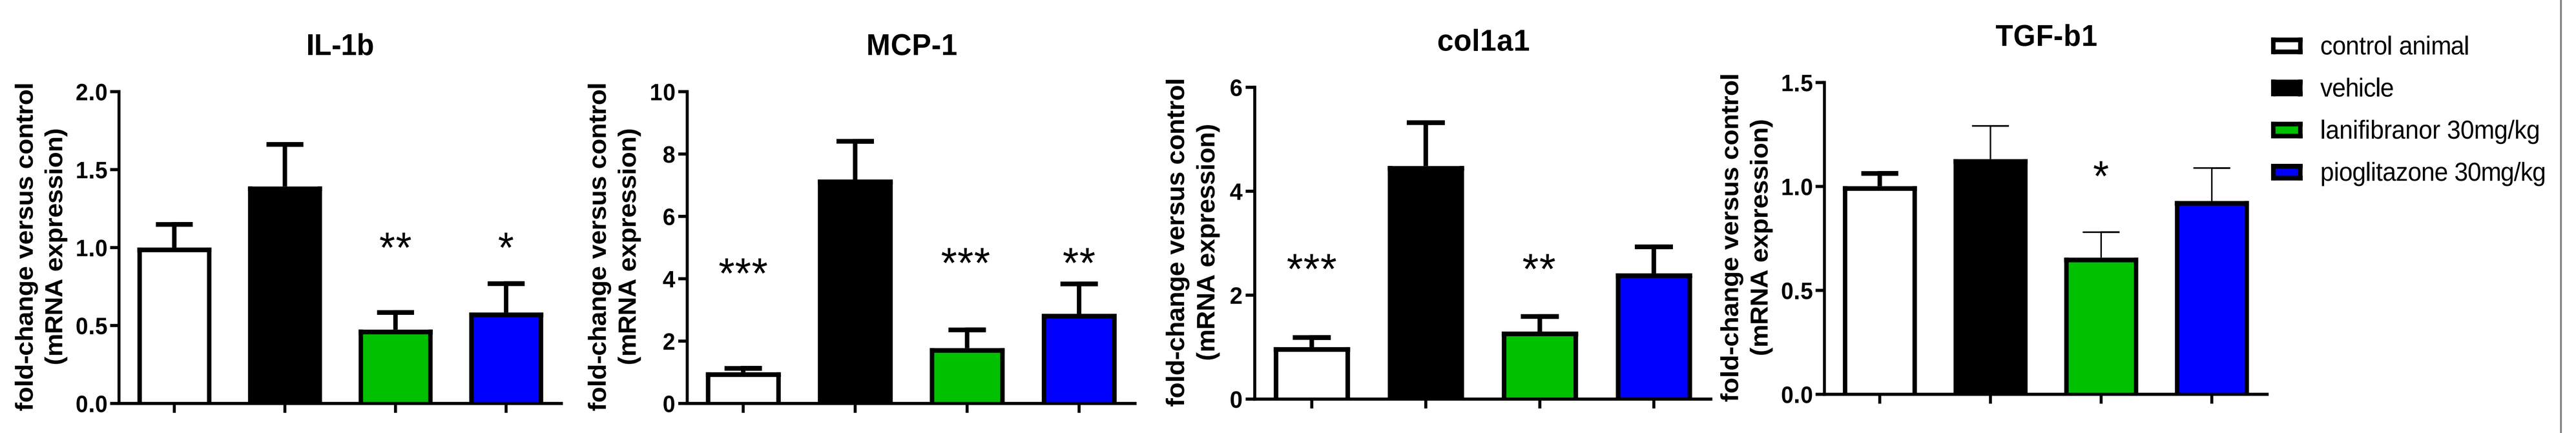
IL-1b
MCP-1
col1a1
TGF-b1
fold-change versus control (mRNA expression)
control animal
vehicle
lanifibranor 30mg/kg
pioglitazone 30mg/kg

Lanifibranor and pioglitazone significantly reduced the expression of inflammatory genes but only lanifibranor significantly reduced the expression of genes associated to fibrosis.

Diastolic dysfunction

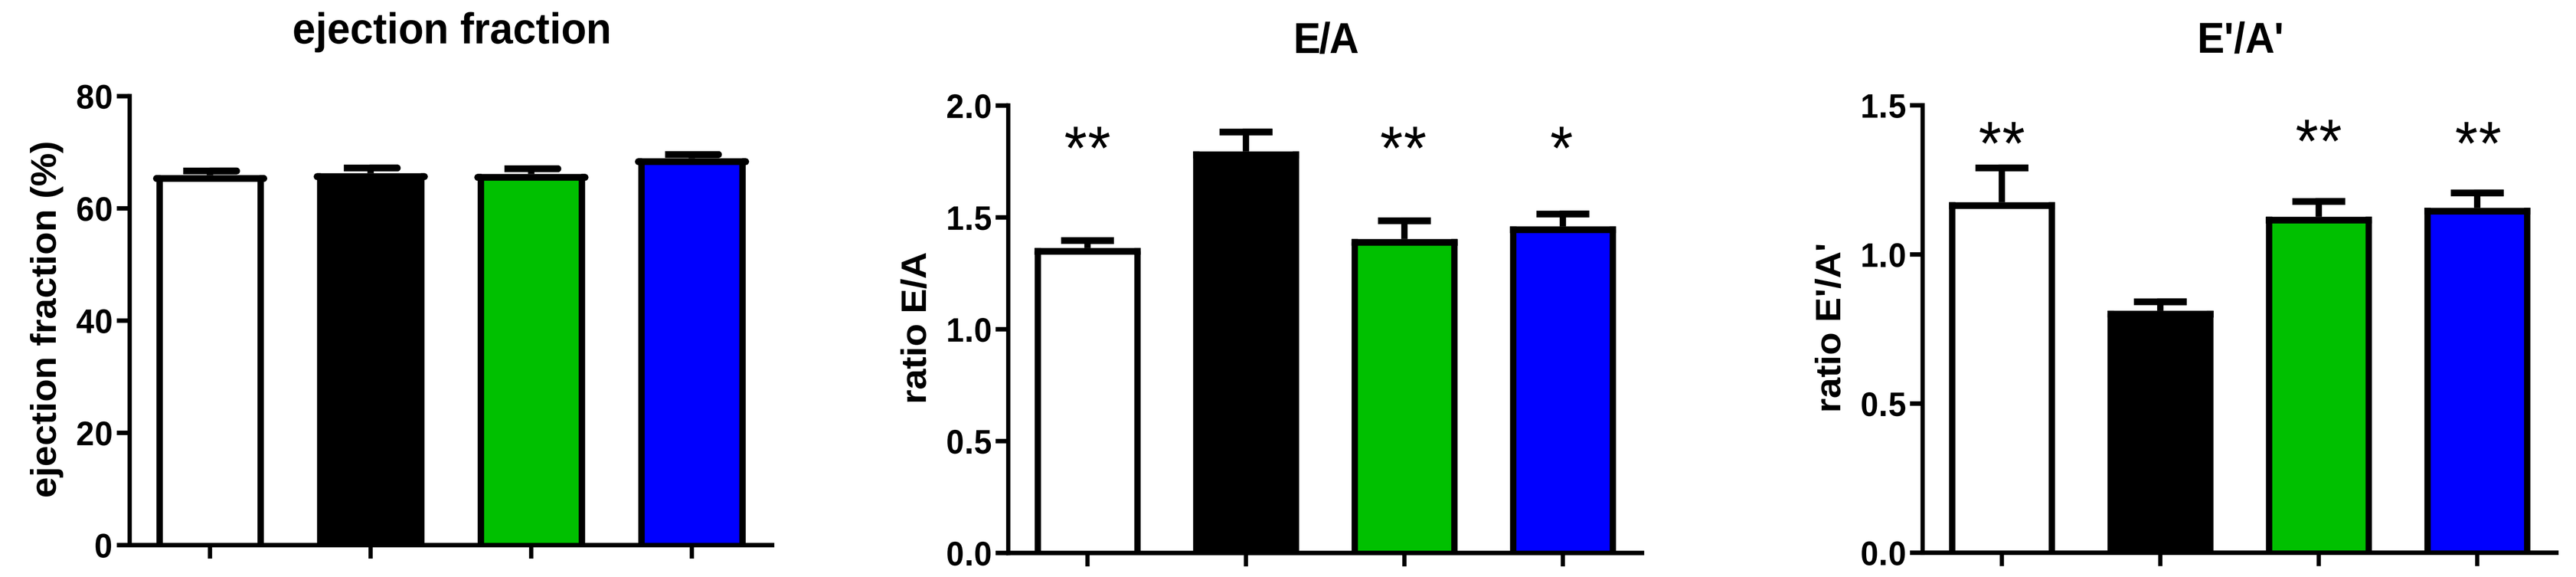
ejection fraction
ejection fraction (%)
E/A
ratio E/A
E'/A'
ratio E'/A'

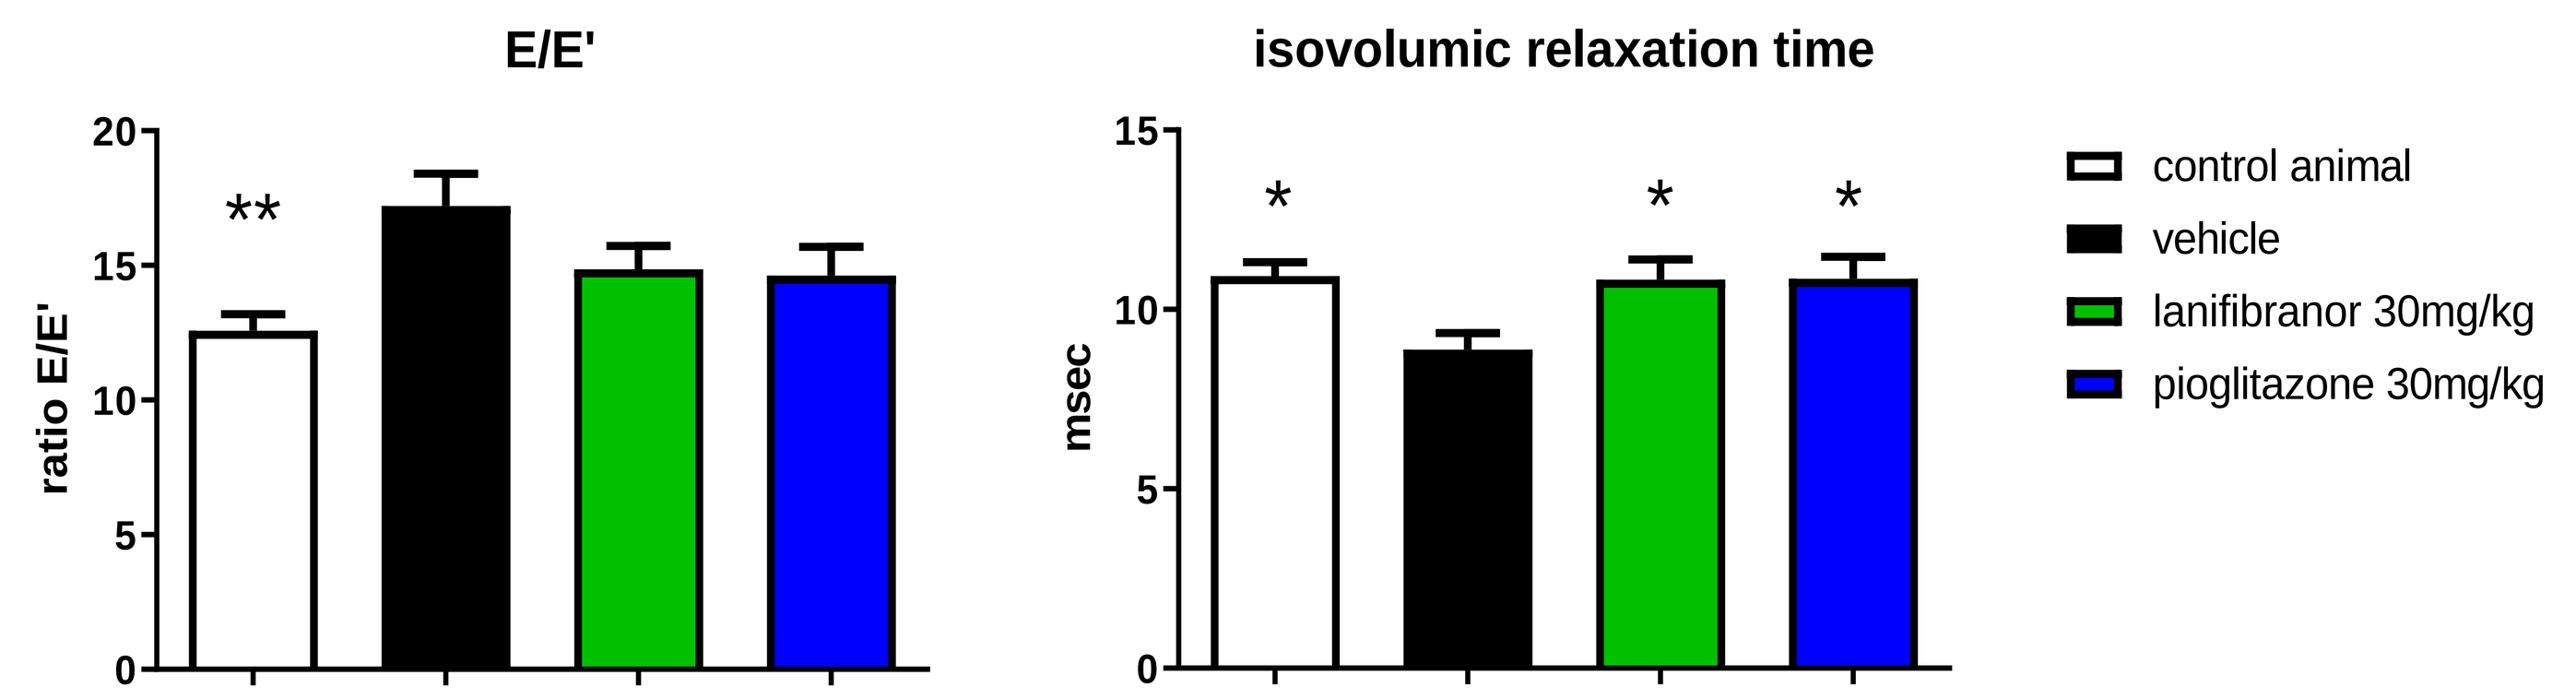
E/E'
ratio E/E'
isovolumic relaxation time
msec
control animal
vehicle
lanifibranor 30mg/kg
pioglitazone 30mg/kg

E/A: Ratios of peak velocities of early to late mitral inflow
E'/A': mitral annular velocities
E/E': a marker of left ventricular filling pressure

Diastolic dysfunction is a cardiac condition associated with left ventricular relaxation or compliance abnormalities and a preserved ejection fraction. High Fat Diet induced an advanced diastolic dysfunction. Lanifibranor and pioglitazone normalized E/A and E'/A' ratio as well as the isovolumic relaxation time and tend to reduced E/E' ratio

4-CONCLUSION

Lanifibranor but not pioglitazone led to improvement in NASH and fibrosis suggesting that a panPPAR activation leads to a greater efficacy than a PPARγ activation alone for the treatment of NASH-related liver features. Lanifibranor also markedly and significantly improved diastolic dysfunction similarly to pioglitazone. Activation of PPARγ is therefore sufficient to correct the diastolic dysfunction in this model. However Lanifibranor as a pan-PPAR activator has the potential to limit the development of diastolic dysfunction via its beneficial effect on NASH but also by a direct effect on diastolic dysfunction through its PPARγ component. This new data further support the development of lanifibranor as a treatment for patients with NASH who are at risk for cardiovascular diseases.

Contact information

Dr. Guillaume Wettstein
Guillaume.wettstein@inventivapharma.com