Combination therapy of lanifibranor and firsocostat further improves steatohepatitis and fibrosis compared to monotherapy in a diet-induced murine model of NASH

Authors, G. WETTSTEIN [1], F. BRIAND [2], T. SULPICE [2], J-L. JUNIEN [1], P. BROQUA [1]

1 Inventiva, Daix, France

2 Physiogenex, Escalquens, France

INTRODUCTION

Lanifibranor, a panPPAR agonist showed clinical efficacy on both resolution of NASH and fibrosis improvement in patients with NASH (NATIVE study). Firsocostat, an ACC1/2 inhibitor, reduced liver fat and markers of fibrosis in patients with advanced fibrosis due to NASH. ACC inhibitors inhibit de novo lipogenesis and reactivates beta oxidation, whilst PPARs stimulate lipid catabolism and redirect hepatic lipids to adipocytes. The potential complementary mode of action of both compounds provides a rationale to investigate whether their combination has additive effects in a nonclinical model of NASH and fibrosis.

METHOD

Mice were fed a 60% high fat/1.25% cholesterol/0.5% cholic acid diet with 2% 2-hydroxypropyl beta-cyclodextrin in drinking water for 3 weeks. After 1 week of diet, mice were randomized according to their ALT, AST plasma levels and body weight and were orally treated (QD) for 2 weeks either with vehicle, lanifibranor (suboptimal dose; 10 mg/kg), firsocostat (15 mg/kg) or the combination lanifibranor and firsocostat. Biochemistry, liver histology and gene expression were analyzed.

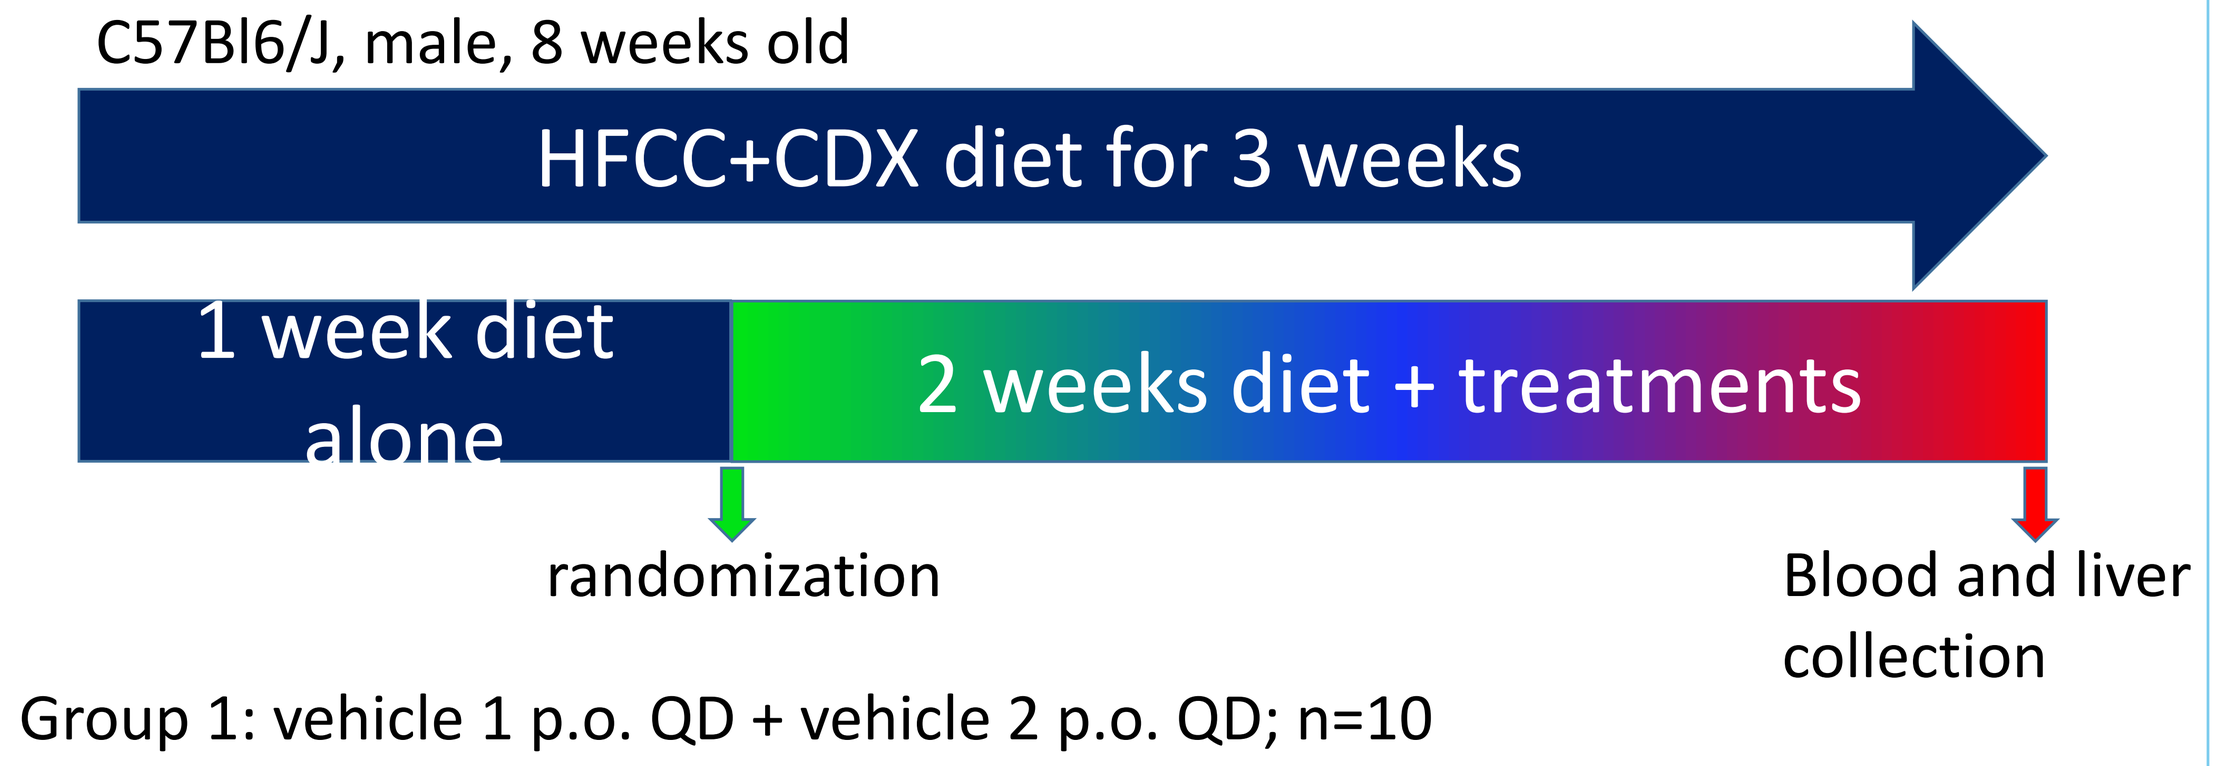
C57Bl6/J, male, 8 weeks old
HFCC+CDX diet for 3 weeks
1 week diet alone
2 weeks diet + treatments
randomization
Blood and liver collection

Group 1: vehicle 1 p.o. QD + vehicle 2 p.o. QD; n=10
Group 2: lanifibranor p.o. QD + vehicle 2 p.o. QD; n=10
Group 3: vehicle 1 p.o. QD + firsocostat p.o. QD; n=10
Group 4: lanifibranor p.o. QD + firsocostat 2 p.o. QD; n=10

RESULTS

* Vs vehicle with * p<0.05; **p<0.01; ***p<0.001; ****p<0.0001
§ Vs combination lani/firso with § p<0.05; §§p<0.01; §§§p<0.001; §§§§p<0.0001

Pilot study in this model was conducted to evaluate dose-response of lanifibranor. We chose for the combination study the dose of 10 mg/kg as a suboptimal dose for lanifibranor. 30 mg/kg being the optimal dose in this model with substantial and significant reduction of steatosis, inflammation and fibrosis.

Effect of lanifibranor, firsocostat and combination on body weight

body weight follow up during treatment

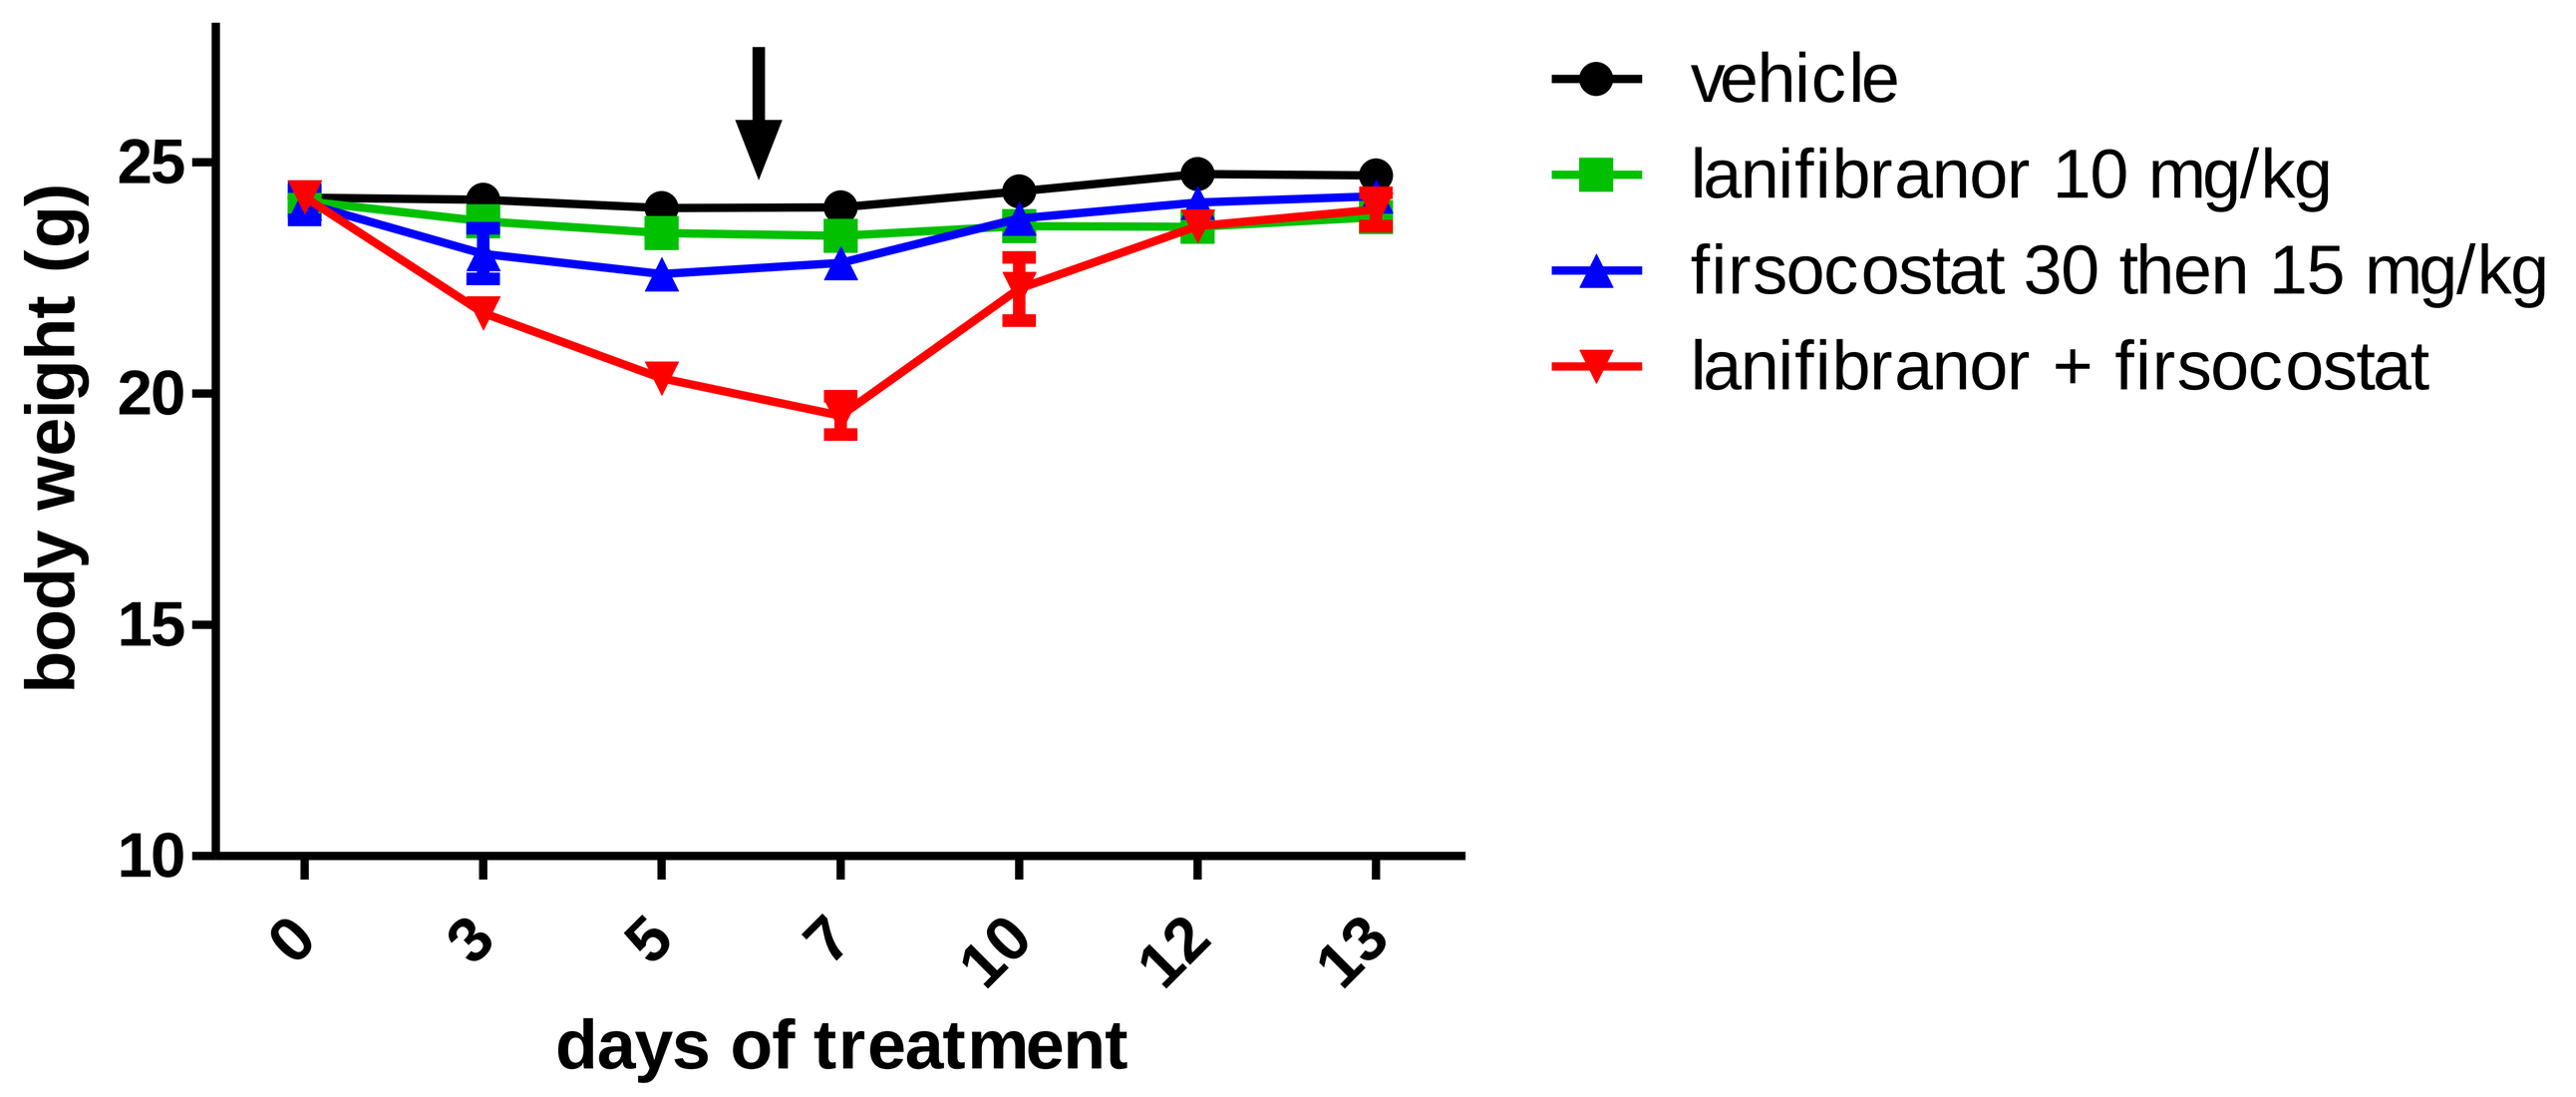
body weight (g)
days of treatment
vehicle
lanifibranor 10 mg/kg
firsocostat 30 then 15 mg/kg
lanifibranor + firsocostat

Lanifibranor had no effect on body weight. Firsocostat was initially given at 30mg/kg but induced body weight lost alone and a drastic body weight decrease in combination with lanifibranor. At day 6 the dose was change to 15 mg/kg in the firsocostat group as well as in the combination group.

Combination therapy further decreases hepatic lipids content

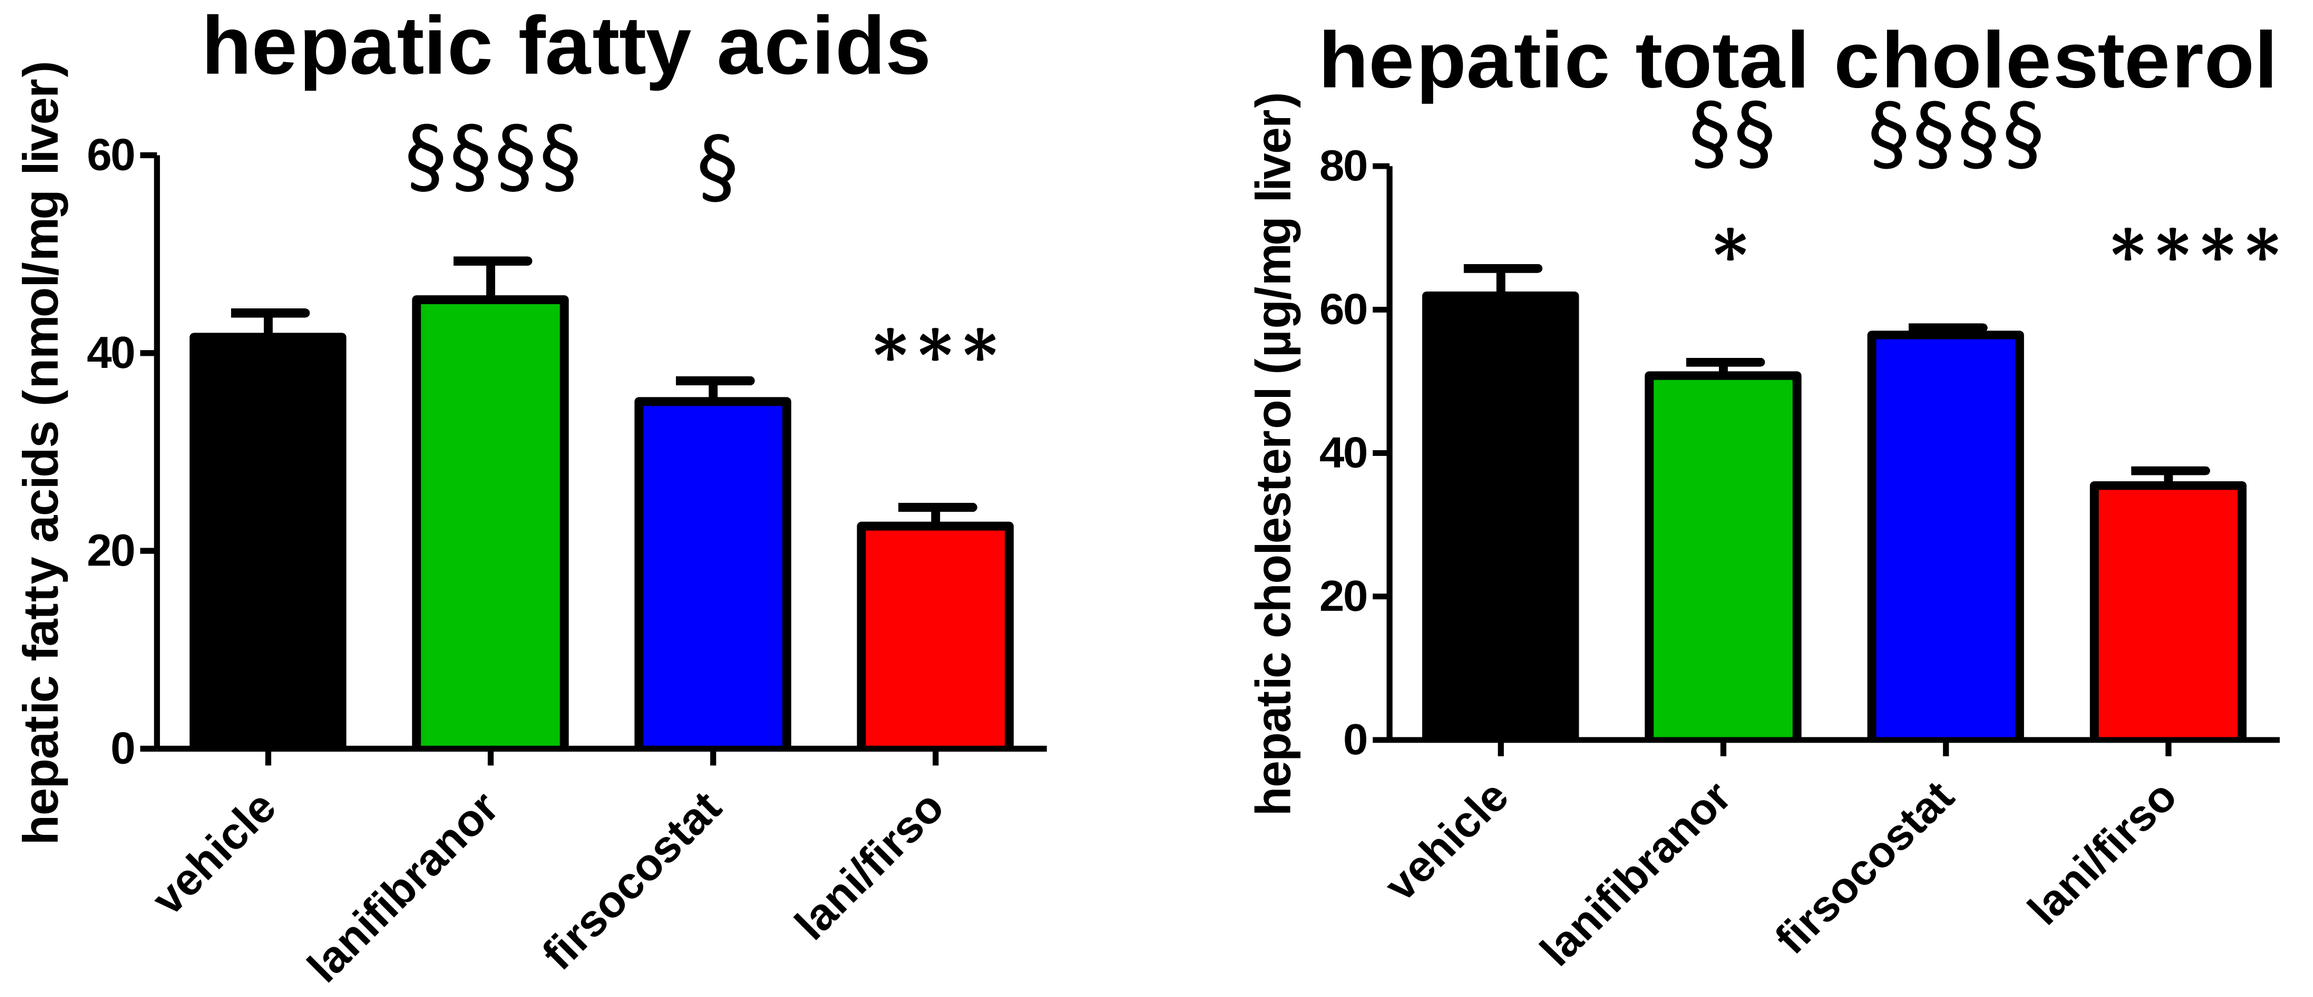
hepatic fatty acids
hepatic fatty acids (nmol/mg liver)
hepatic total cholesterol
hepatic cholesterol (µg/mg liver)
vehicle
lanifibranor
firsocostat
lani/firso

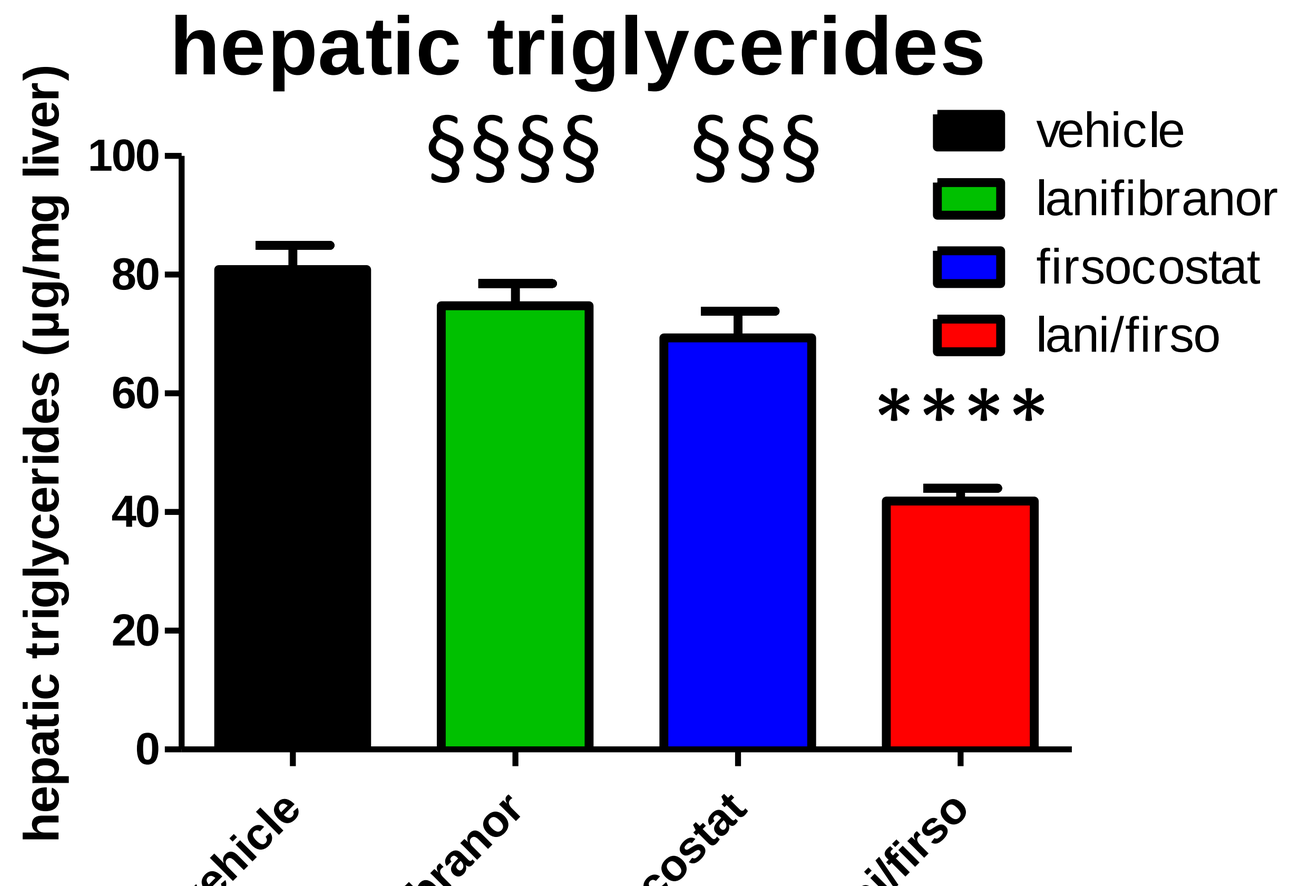
hepatic triglycerides
hepatic triglycerides (µg/mg liver)
vehicle
lanifibranor
firsocostat
lani/firso

In this condition neither lanifibranor nor firsocostat alone had an effect on hepatic lipids content (except for lanifibranor on total cholesterol). However the combination of the two drugs lead to a significant reduction of hepatic lipids content.

Combination therapy strenghten the effect of the two compounds on steatosis and inflammation

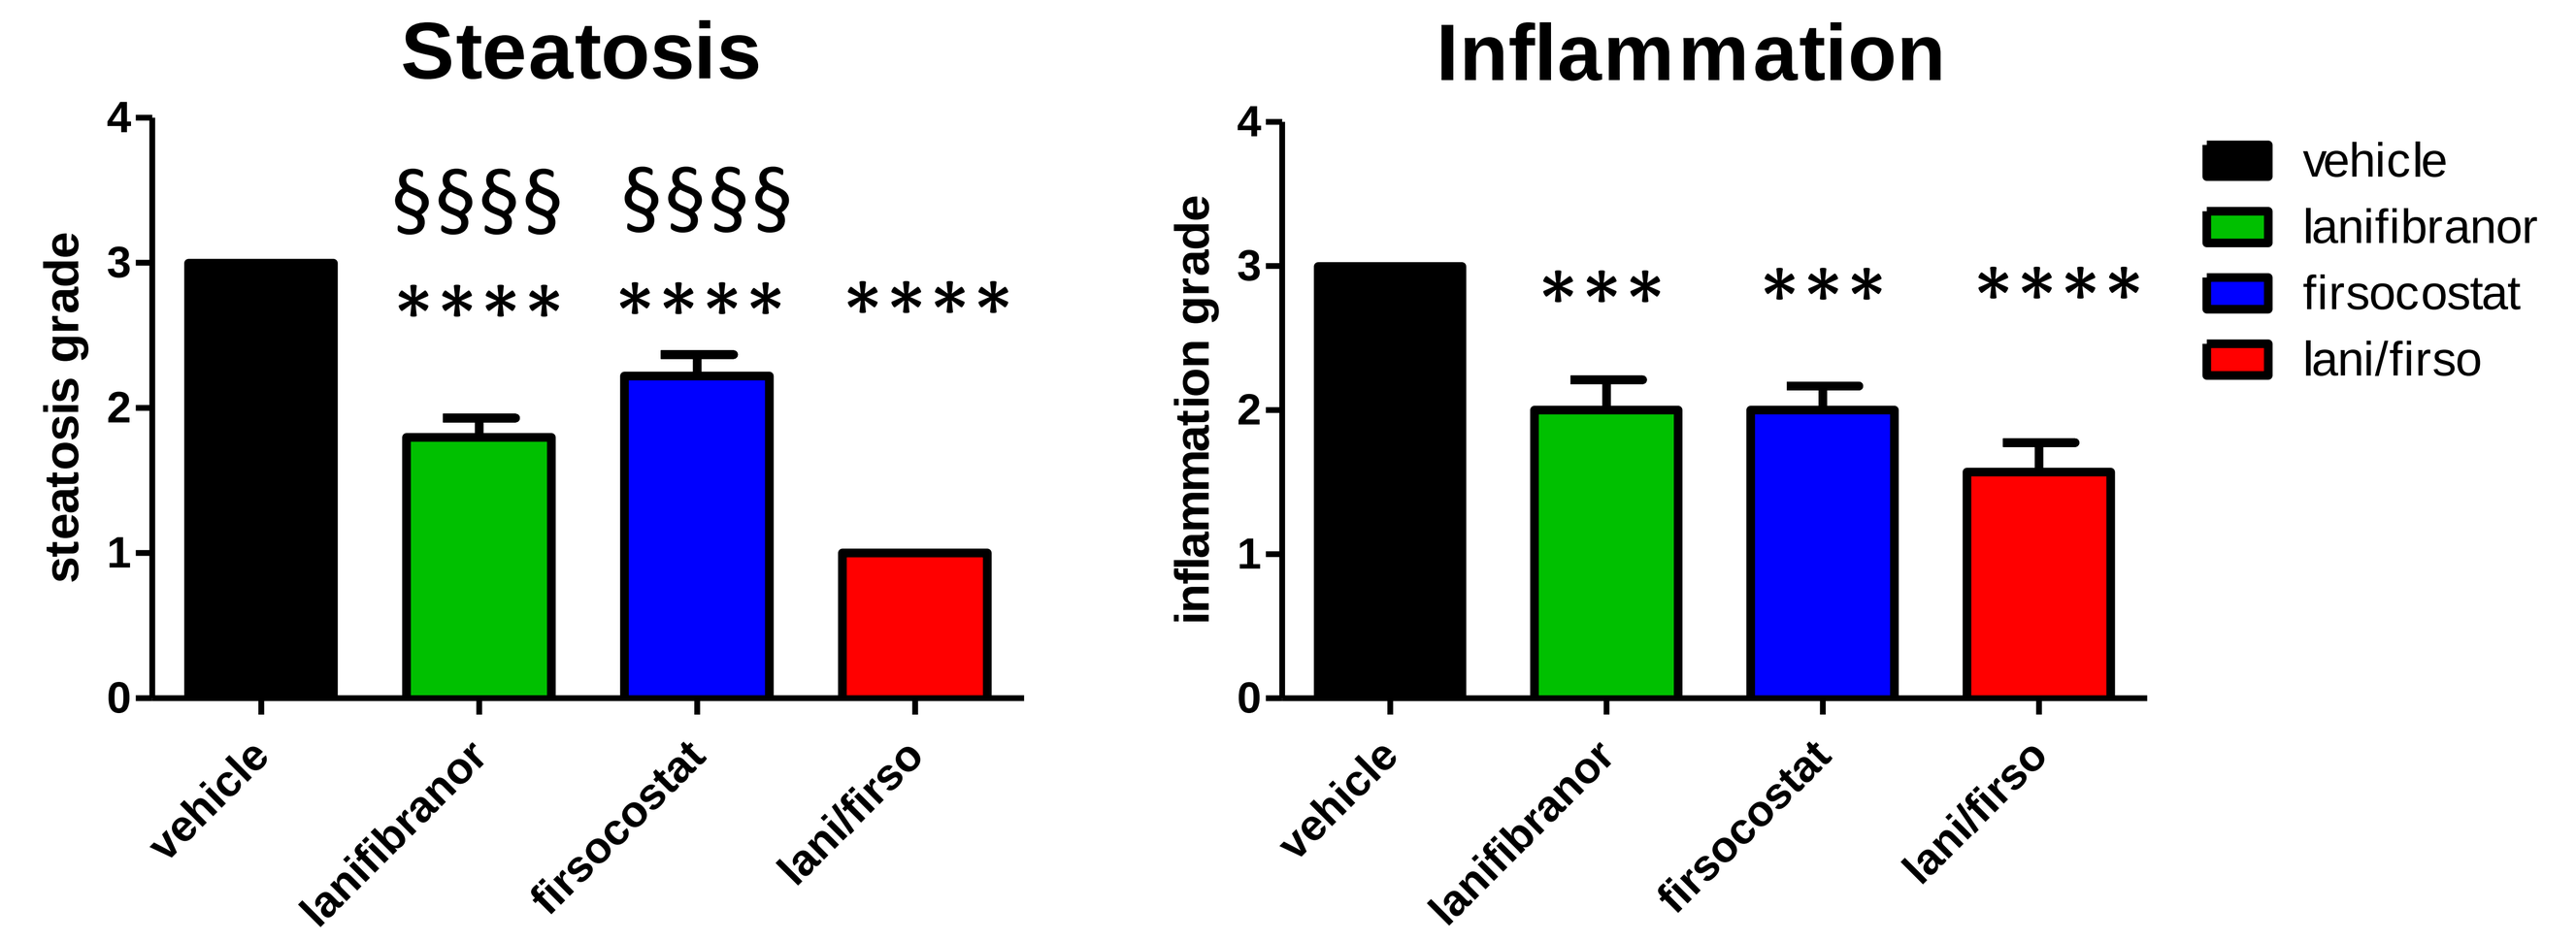
Steatosis
steatosis grade
Inflammation
inflammation grade
vehicle
lanifibranor
firsocostat
lani/firso

Lanifibranor and firsocostat alone already reduced steatosis and inflammation and combination therapy improved significantly the effect on steatosis.

Combination therapy inhibits fibrosis

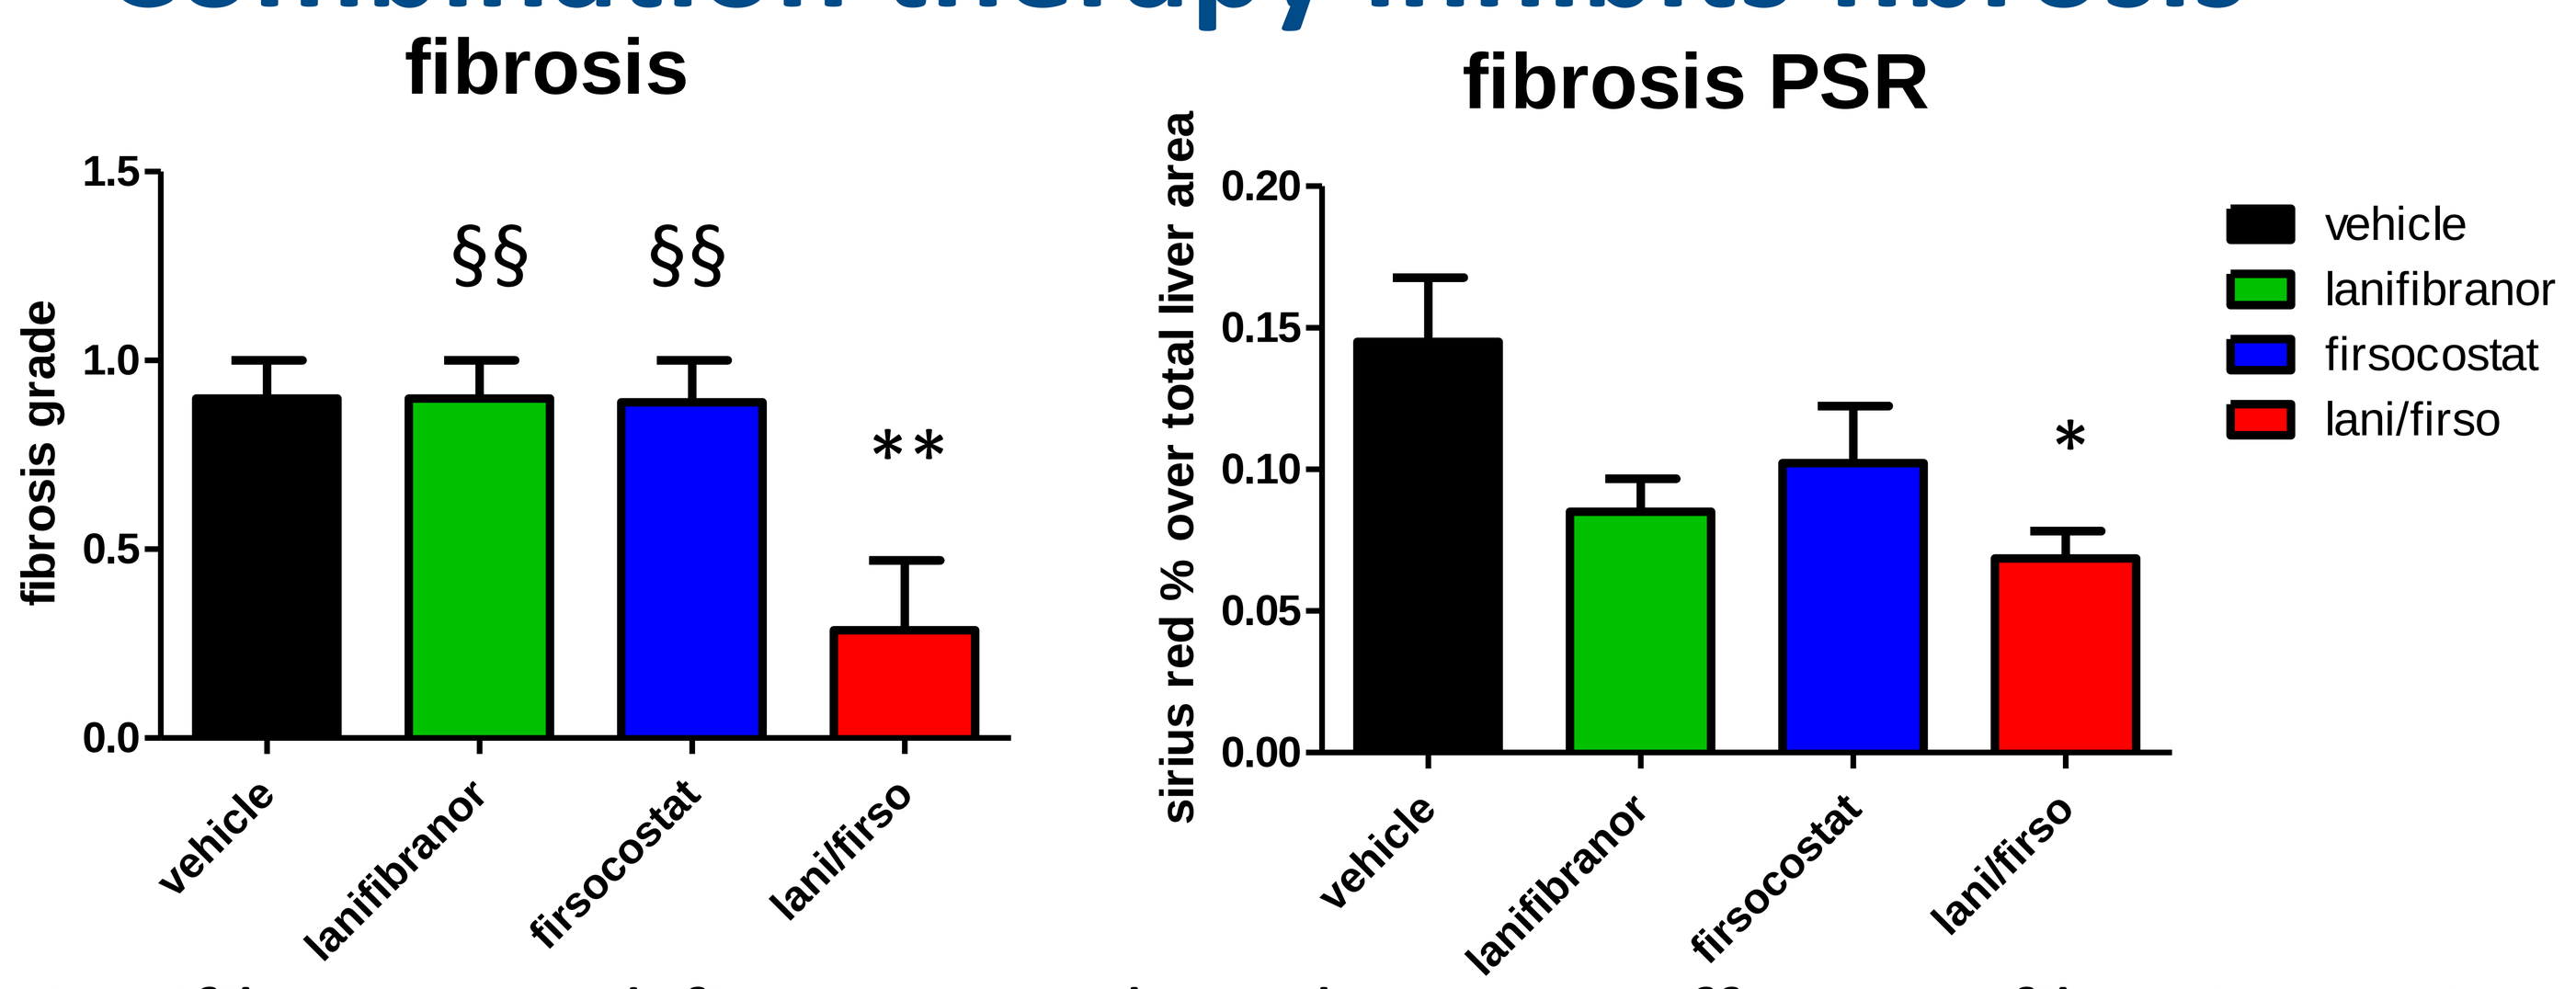
fibrosis
fibrosis grade
fibrosis PSR
sirius red % over total liver area
vehicle
lanifibranor
firsocostat
lani/firso

Lanifibranor and firsocostat alone have no effect on fibrosis scoring but the combination lead to markedly and significant reduction of fibrosis. However when looking at collagen deposition (PSR) we observed a trend of decrease with lanifibranor. The combination therapy being thus significant vs vehicle but not vs the two compounds

Combination therapy further improves total scoring

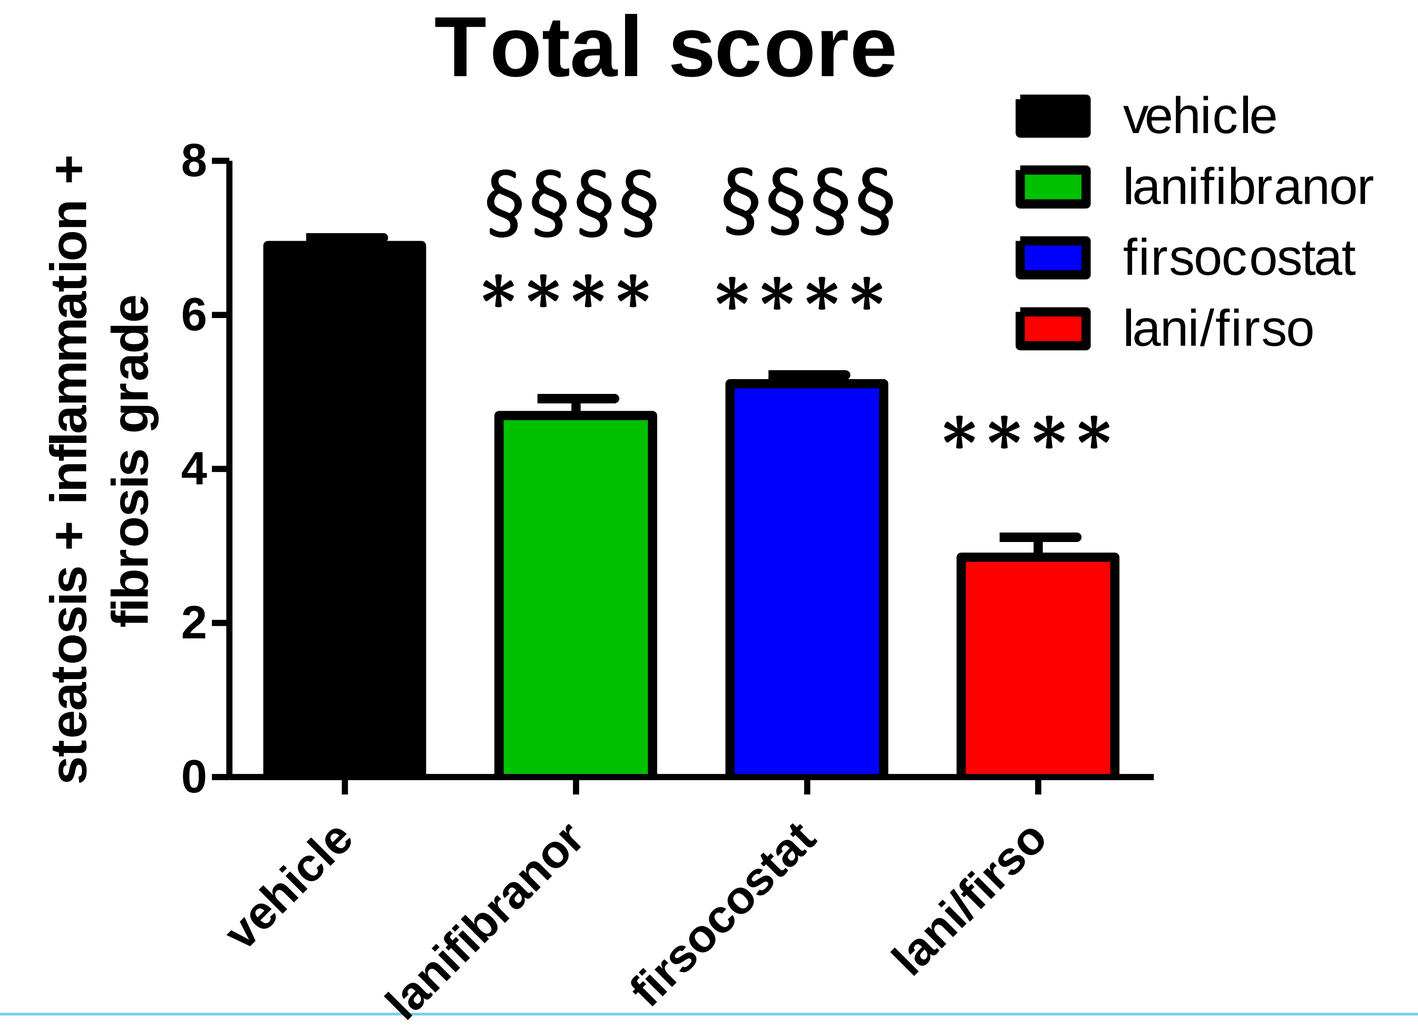
Total score
steatosis + inflammation + fibrosis grade
vehicle
lanifibranor
firsocostat
lani/firso

Lanifibranor and firsocostat alone significantly improve the total scoring (steatosis, inflammation and fibrosis) through their beneficial effect on steatosis and inflammation. The combination further improves the total score due to the improvement observed on steatosis and fibrosis scoring.

Combination results in a more efficient effect on Col1a1 and TGF-β1

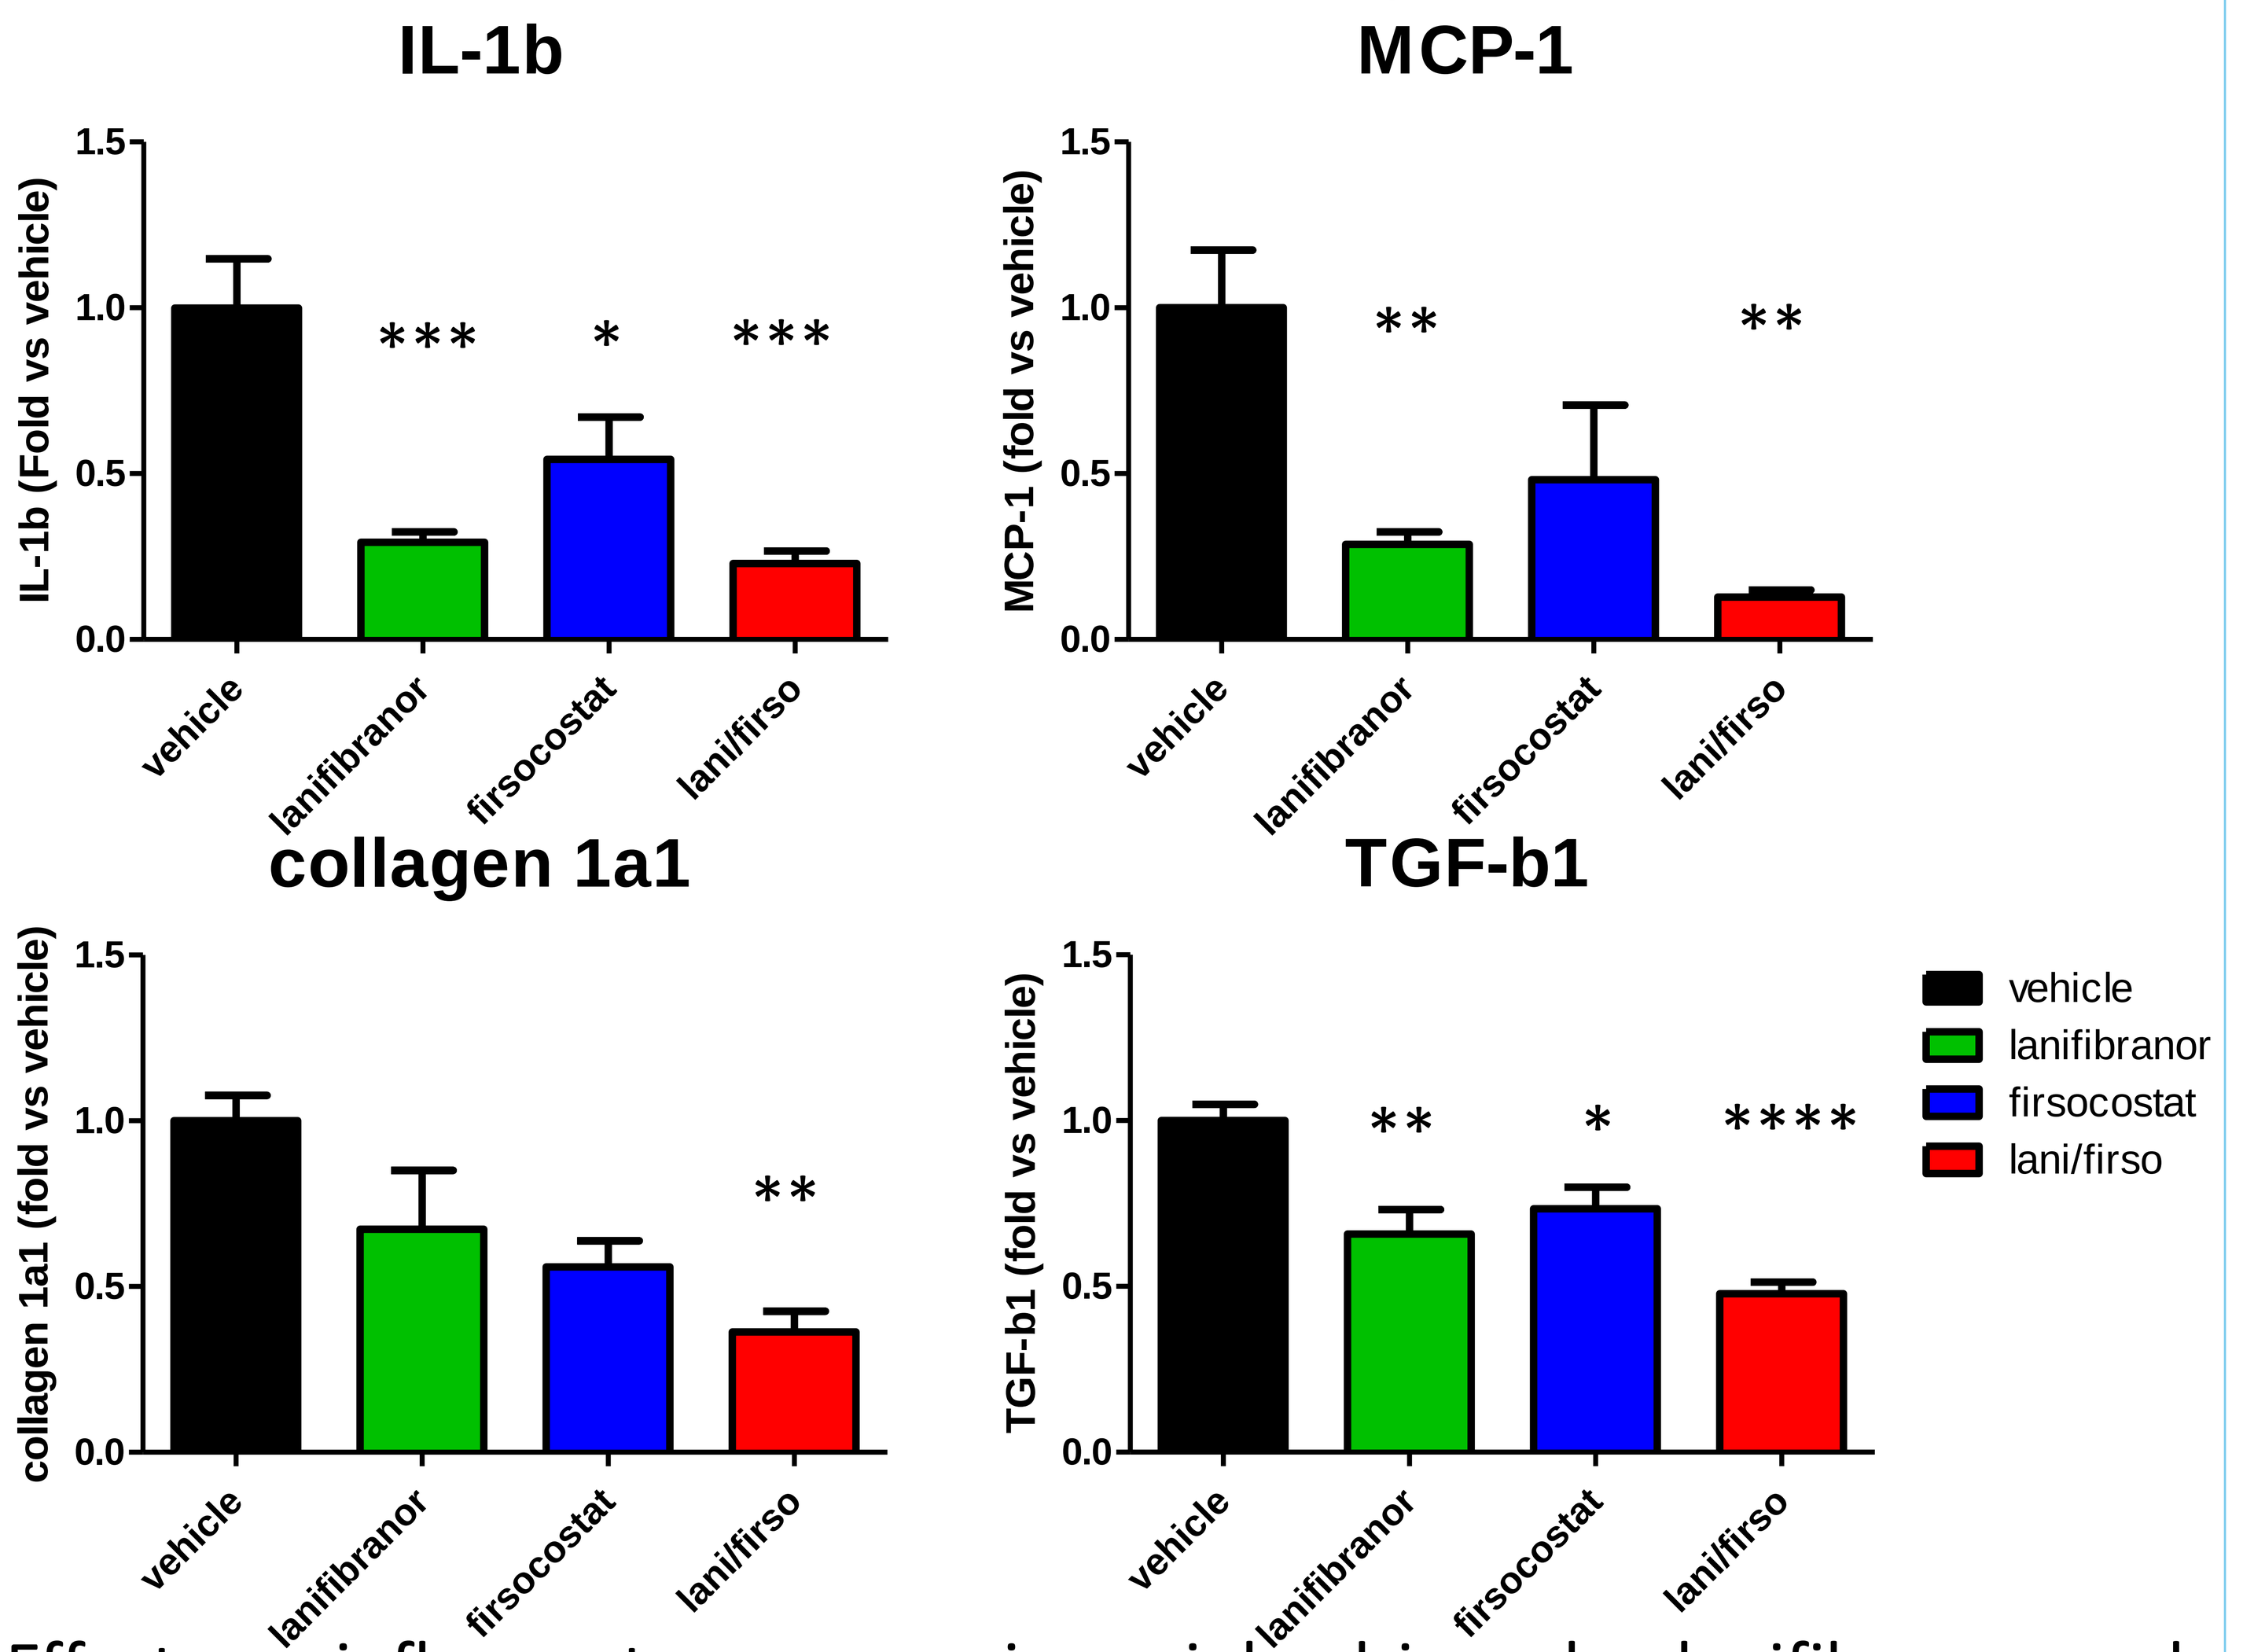
IL-1b
IL-1b (Fold vs vehicle)
MCP-1
MCP-1 (fold vs vehicle)
collagen 1a1
collagen 1a1 (fold vs vehicle)
TGF-b1
TGF-b1 (fold vs vehicle)
vehicle
lanifibranor
firsocostat
lani/firso

Effect on inflammatory genes is mainly driven by lanifibranor and combination therapy did not result in a statistical improvement. Lanifibranor and firsocostat demonstrated a trend of improvement and an improvement on collagen 1a1 and TGF-β1 respectively. The combination improved the statistical significance vs vehicle but did not result in a statistical improvement vs the two compounds

CONCLUSIONS

Lanifibranor and firsocostat combination reached greater efficacy than monotherapy at the selected doses on hepatic lipid content, steatosis, fibrosis and total scoring. These data emphasize the complementary effect of these two compounds on lipids metabolic leading to further improvement of NASH and fibrosis. These data would support clinical investigation of a combination of lanifibranor and firsocostat in patients with NASH.

CONTACT INFORMATION

Guillaume.wettstein@inventivapharma.com